Evaluation of Brilaroxazine (RP5063) in a Bleomycin-Induced Rodent Model of Idiopathic Pulmonary Fibrosis

Laxminarayan Bhat[a], Seema R Bhat[a], Marie-Claude Nault[b], Marzena Biernat[b], Sebastien M. Labbe[b]

[a]Reviva Pharmaceuticals Holdings, Inc., 19925 Stevens Creek Blvd, Suite 100, Cupertino, California 95014, USA

[b]IPS Therapeutique 3035 Boulevard Industriel, Sherbrooke Québec J1L 2T9, CANADA

Poster# 623

INTRODUCTION

Idiopathic pulmonary fibrosis (IPF) is a chronic, progressive, and debilitating lung disease with a global prevalence of ~3 million, 2-5 years in median survival time, and 50,000 annual U.S. deaths[1–6]. It involves chronic inflammation and progressive alveolar fibrosis, leading to destroyed architecture, reduced capacity, impaired oxygenation, and declined function[2,7,8].

Serotonin (5-HT) plays a key role, via 5-$HT_{2A/2B/7}$ receptors, in IPF's pathology involving a vasoactive effect on pulmonary arteries and lung myofibroblast actions[3,9-12].

Brilaroxazine displays a high affinity for $D_{2/3/4}$ and 5-$HT_{2A/2B/7}$ receptors and moderate affinity for the serotonin transporter[13,14]. Its effects on vascular fibrosis (5-HT_{2B} receptor), proliferation (5-$H_{2A/2B}$ receptor), relaxation (5-HT_{2A} receptor), inflammation (5-HT_7 receptor), and pro-inflammatory cytokines have created interest in IPF[15-18].

OBJECTIVE

This study evaluated whether brilaroxazine (15 mg twice-daily), started on Day 1 or Day 10, displays efficacy in a bleomycin (BLM)-induced rat model of IPF?

METHODS

The study scheme[19] (Figure 1) involved.

- Day 0: Four groups of Sprague Dawley rats received BLM-induction, and one placebo (Sham) (n=5).
- Day 1: One group started on brilaroxazine 15 mg twice daily (BT) (n=10). Two groups continued on vehicle (BLM) (n=9,10).
- Day 10: One group (n=10) started on brilaroxazine 15 mg twice daily (BI) and one continued on BLM (n=9). All groups continued until Day 20.
- Day 21: investigators collected hemodynamic parameters (systemic arterial blood pressure, heart rate, and oxygen saturation) and blood samples. They sacrificed the animals, then collected bronchoalveolar lavage fluid (BALF), lung, trachea, and heart tissue.

FIGURE 1

Schedule of various treatments of the animals during the bleomycin-induced IPF study

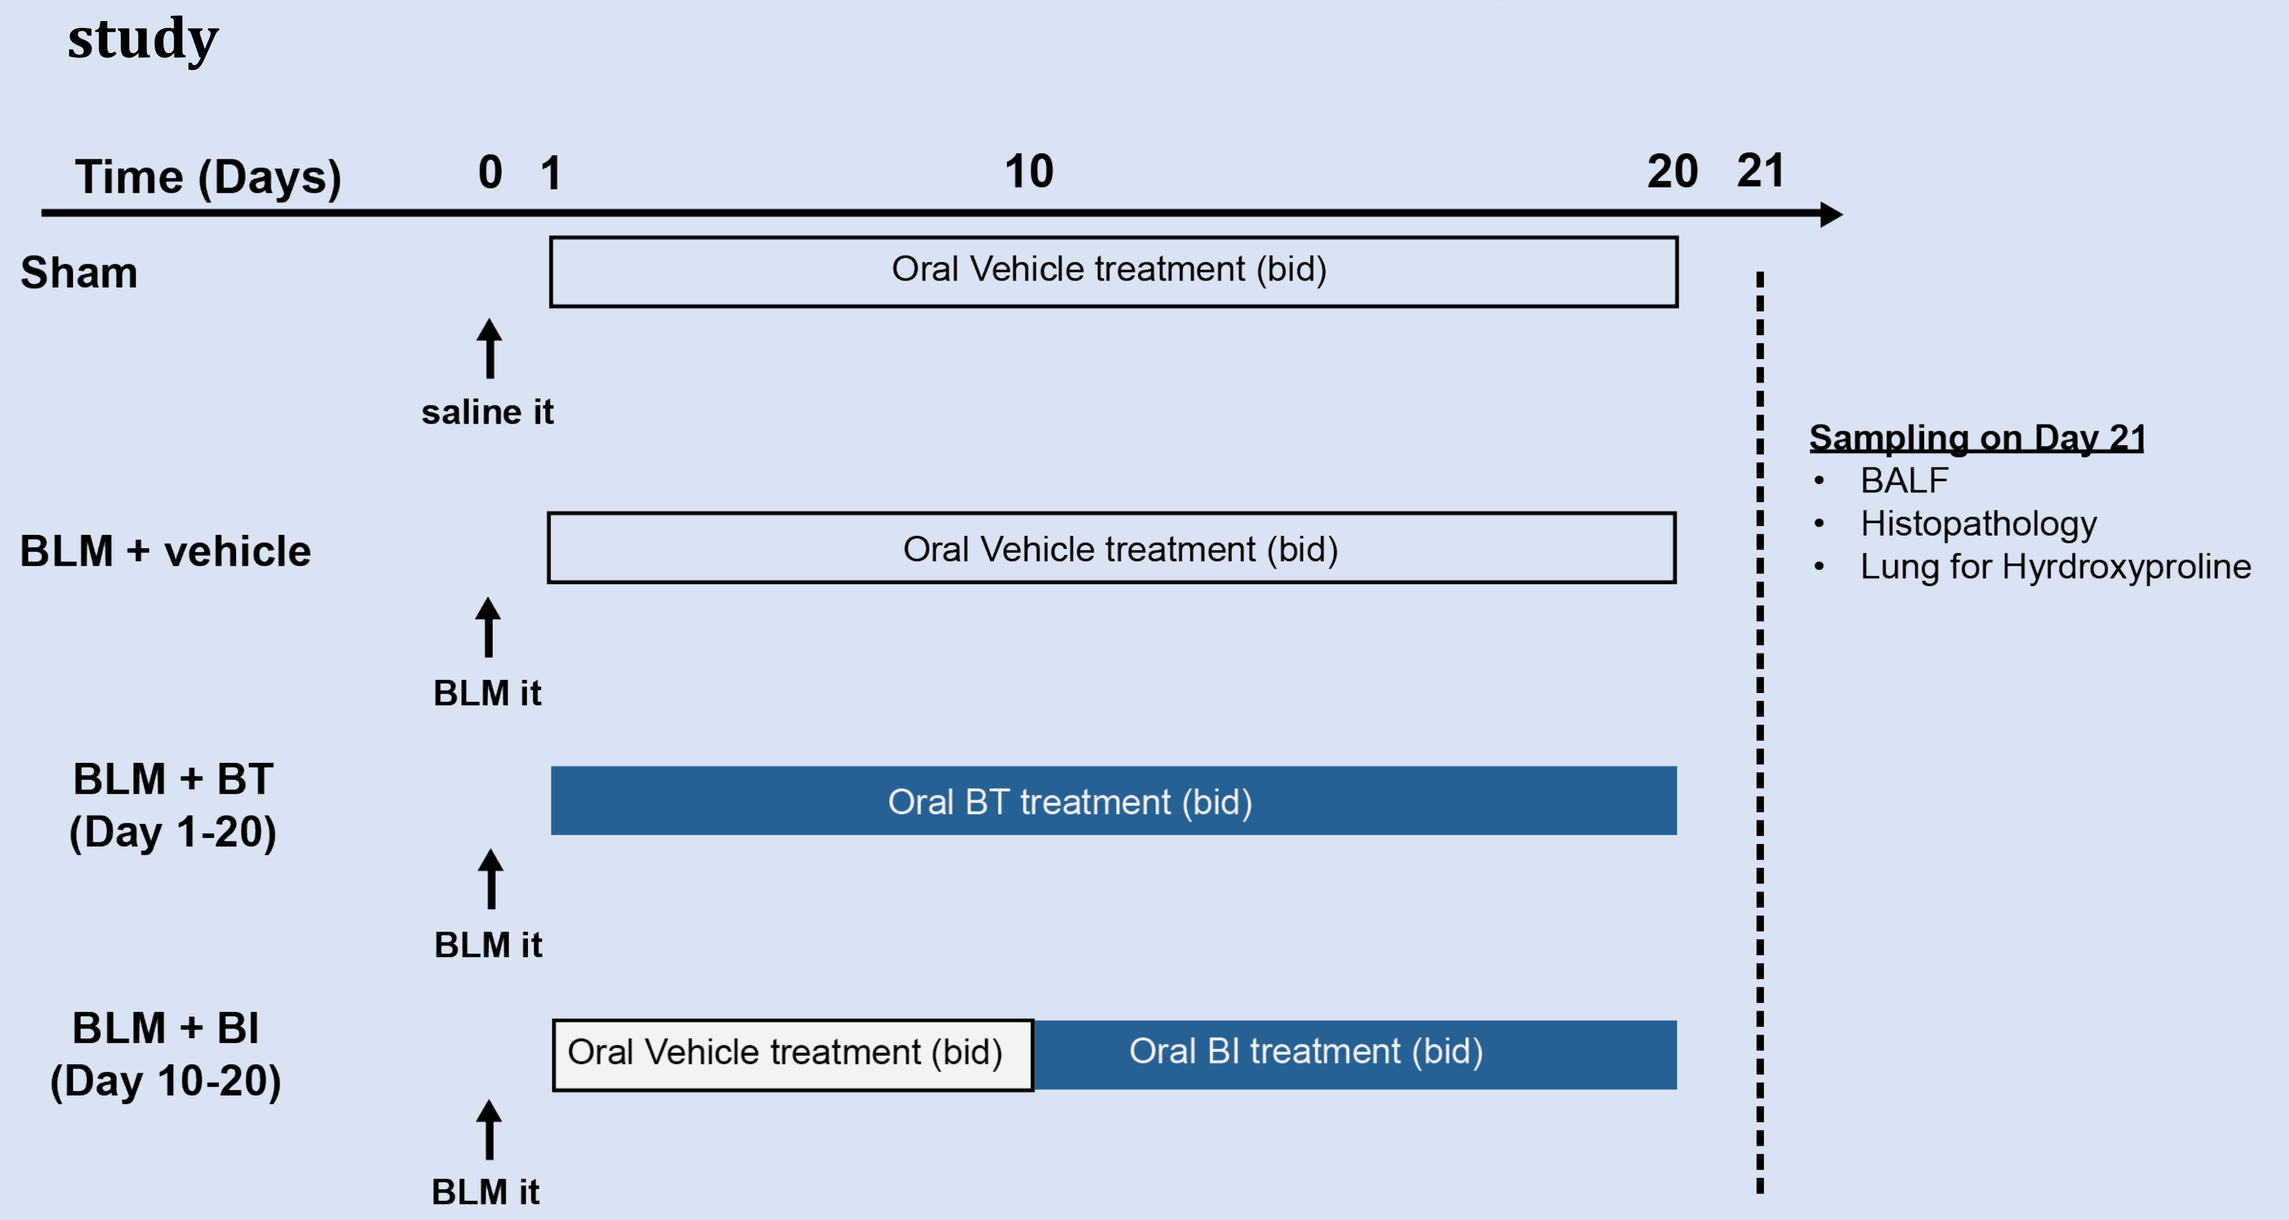
Time (Days) 0 1 10 20 21
Sham
Oral Vehicle treatment (bid)
saline it
BLM + vehicle
Oral Vehicle treatment (bid)
BLM it
BLM + BT (Day 1-20)
Oral BT treatment (bid)
BLM it
BLM + BI (Day 10-20)
Oral Vehicle treatment (bid)
Oral BI treatment (bid)
BLM it
Sampling on Day 21
BALF
Histopathology
Lung for Hyrdroxyproline

Primary outcomes were survival and weight. Others included cardiopulmonary and pressure parameters at surgery, tissue weights, histologic samples (hematoxylin and eosin [H&E] with Ashcroft score and Masson's trichrome staining), BALF cell counts, hydroxyproline levels, and cytokines (macrophage inflammatory protein 1 [MIP1], monocyte chemoattractant protein 1 [MCP1], Interleukin [IL]-6, Interferon gamma-induced protein 10 [IP10]; and RANTES.

Statistical comparisons involved ANOVA (then a Fisher's exact post hoc test for BLM). Significance was P < 0.05.

RESULTS

FIGURE 2

Survival curves from Day 1-10 (A) and Day 11-21 (B), and body weights (C) of Sham, BLM-induced, and treatment group animals

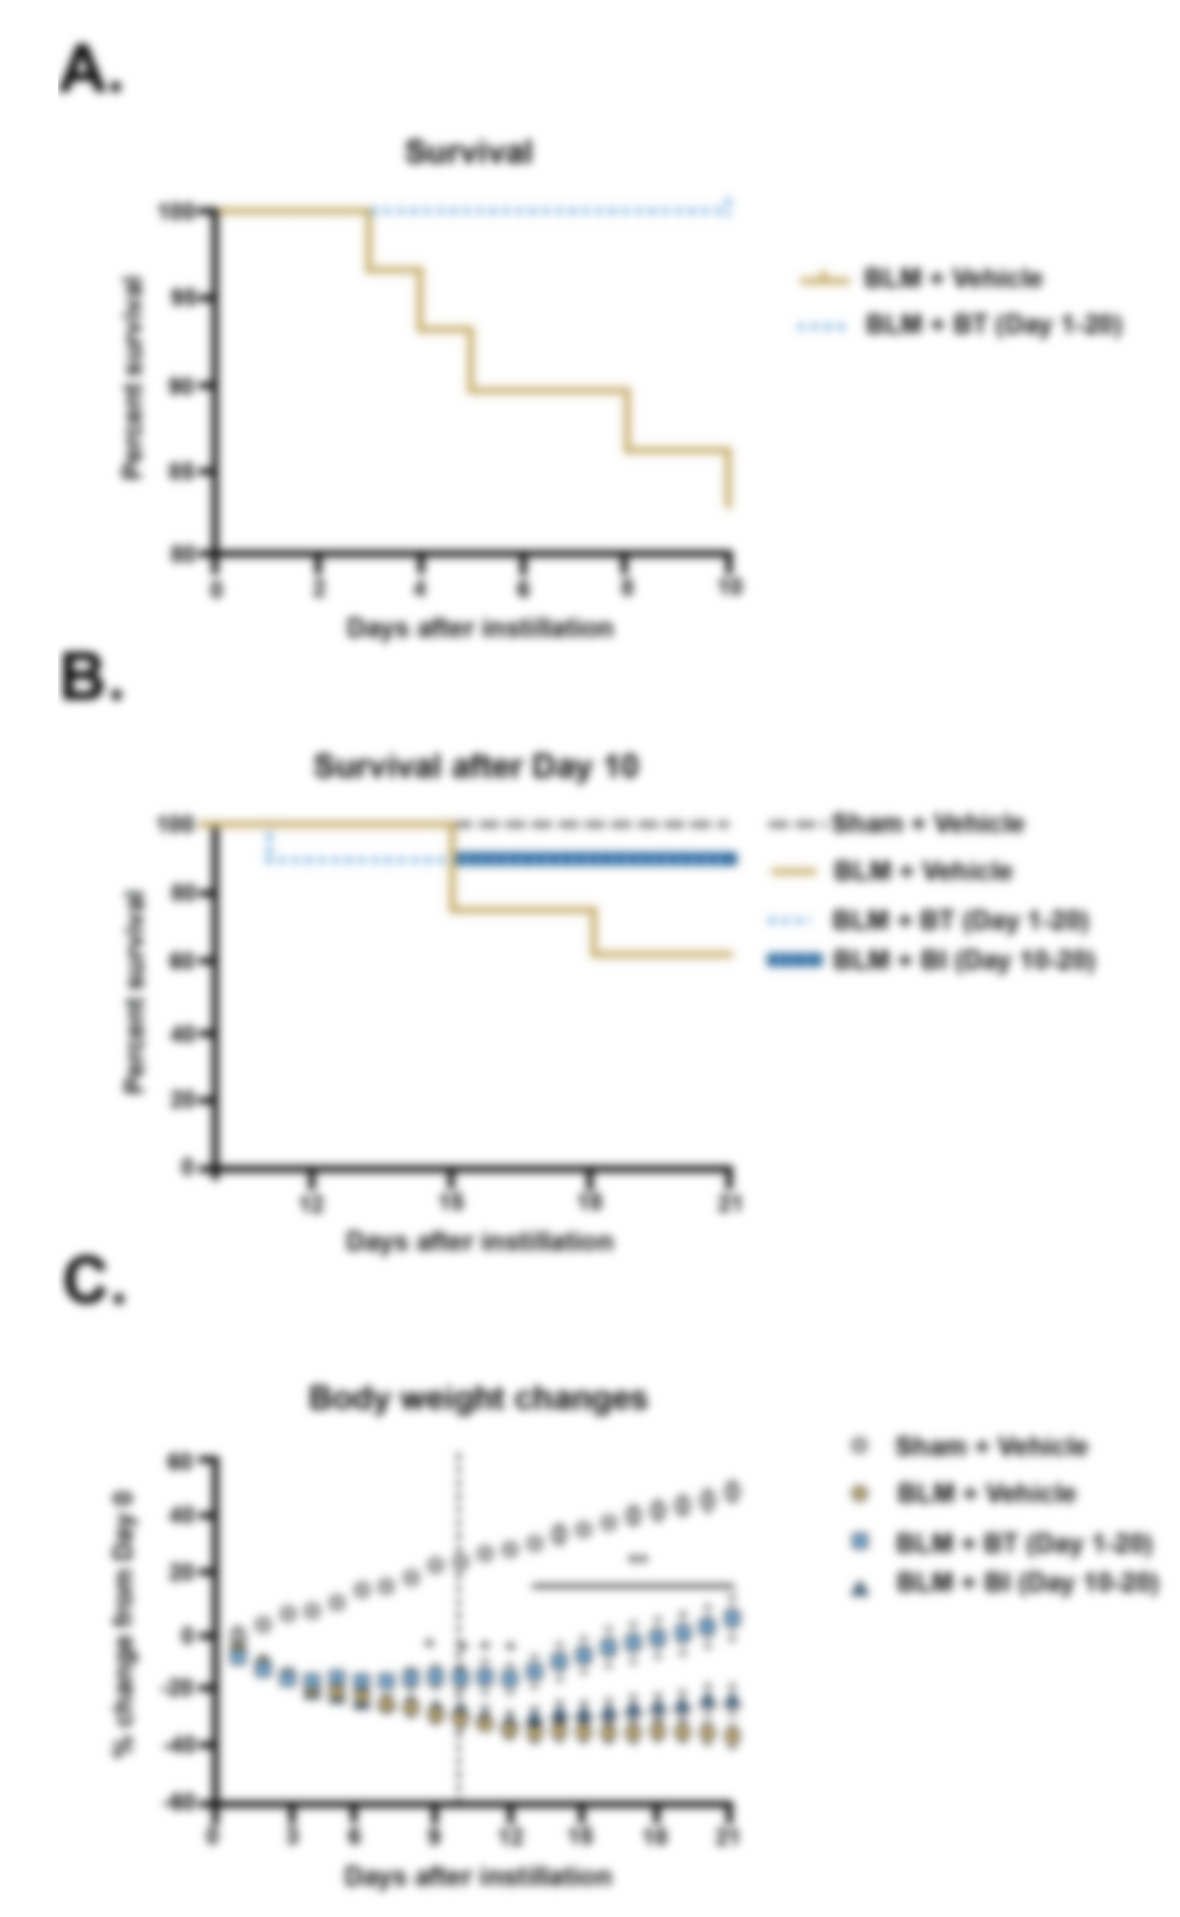
A.
Survival
B.
Survival after Day 10
C.
Body weight changes

BLM: Bleomycin; Sham: Non-induced animals with the vehicle.
*P<0.05 BLM vs. Sham. ## P<0.05, vs. BLM.
Day 21 survival (Figure 2A,B) with BT and BI were 90% and 89.5%, respectively (P<0.05, BLM at 62%). BT significantly alleviated BLM-induced weight loss (P<0.01, BLM) (Figure 2C).

FIGURE 3

Hemodynamic and cardiac parameters (A-C) and systemic arterial pressures (D-F) measured on Day 21

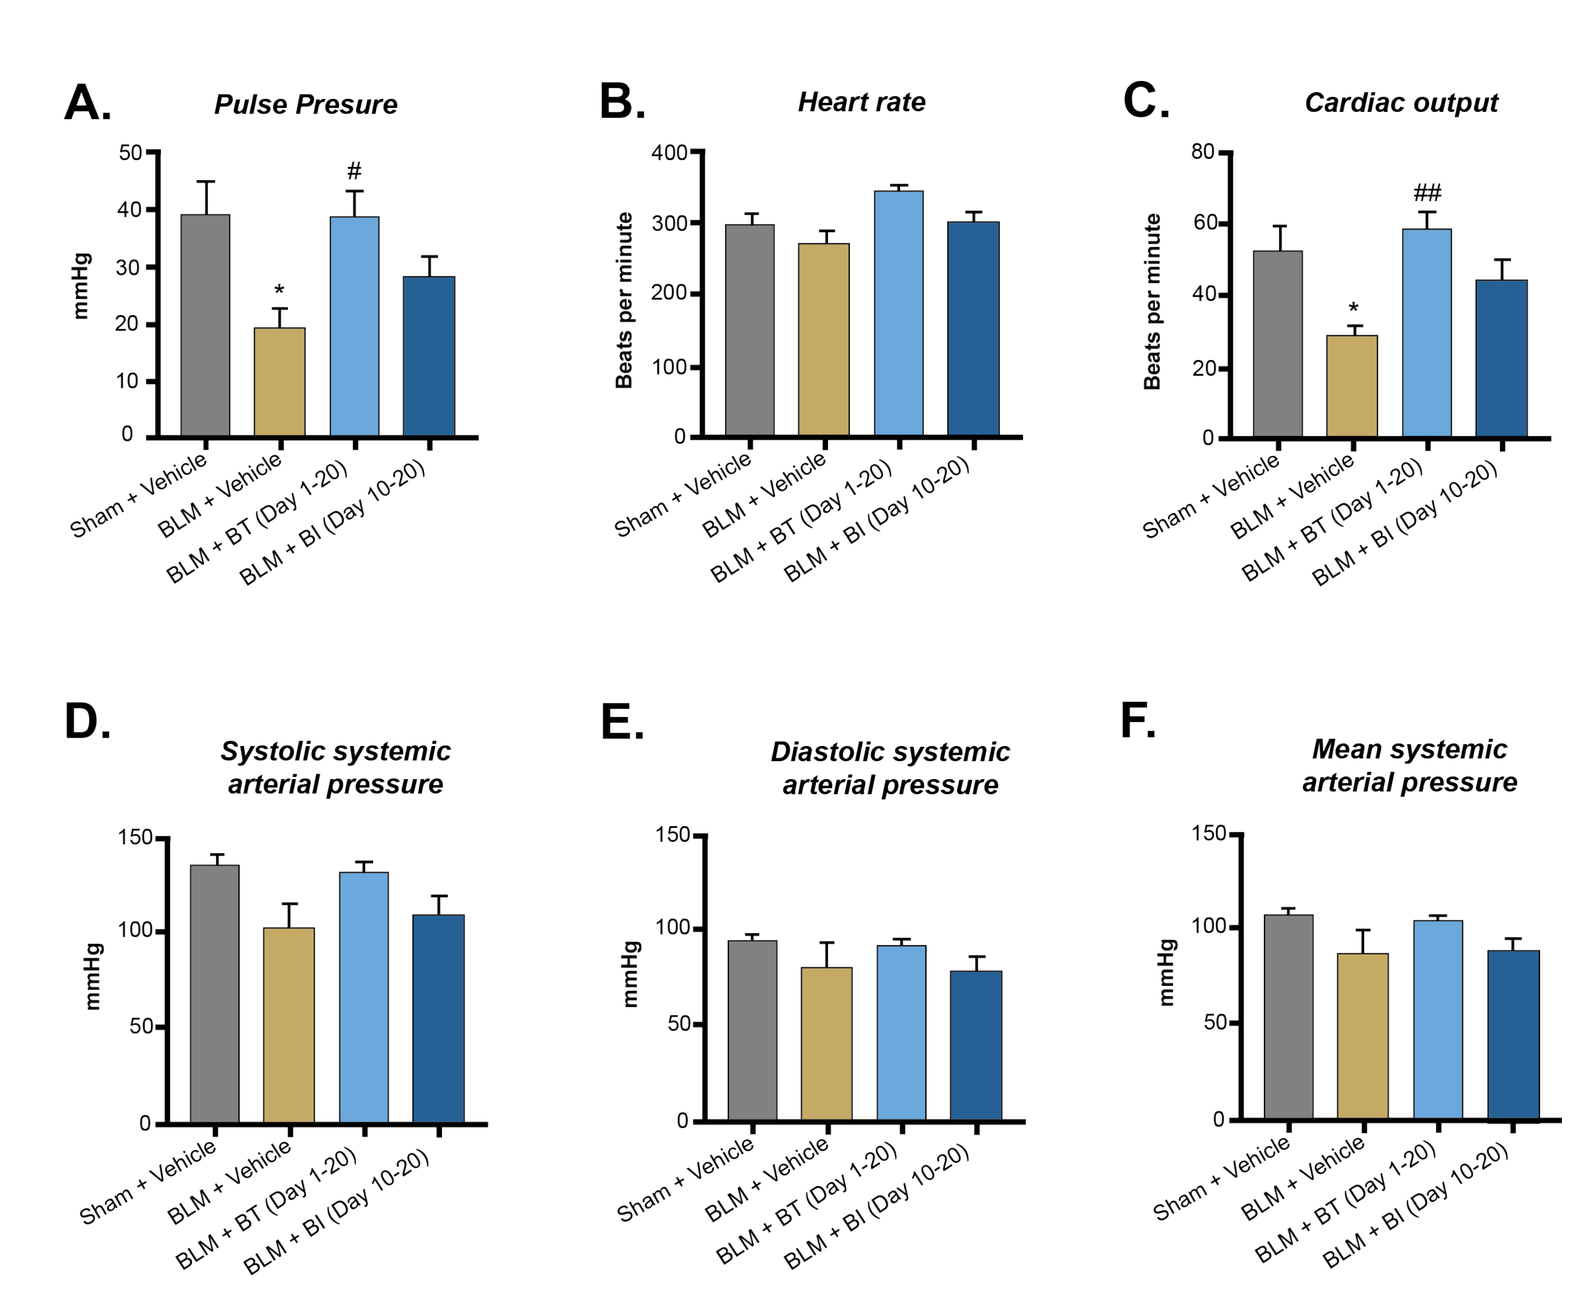
A.
Pulse Presure
B.
Heart rate
C.
Cardiac output
D.
Systolic systemic arterial pressure
E.
Diastolic systemic arterial pressure
F.
Mean systemic arterial pressure

BLM: Bleomycin; Sham: Non-induced animals with the vehicle.
*P<0.05 BLM vs. Sham. ## P<0.05 vs. BLM+Veh.
BT experienced an improved arterial pulse pressure (P<0.05, BLM) (Figure 3A). Both treatments restored cardiac output; BT was significant (P<0.01, BLM) (Figure 3C).

FIGURE 4

BLM-induced effects on blood oxygen saturation (A) and blood lactate levels (B) measured on Day 21

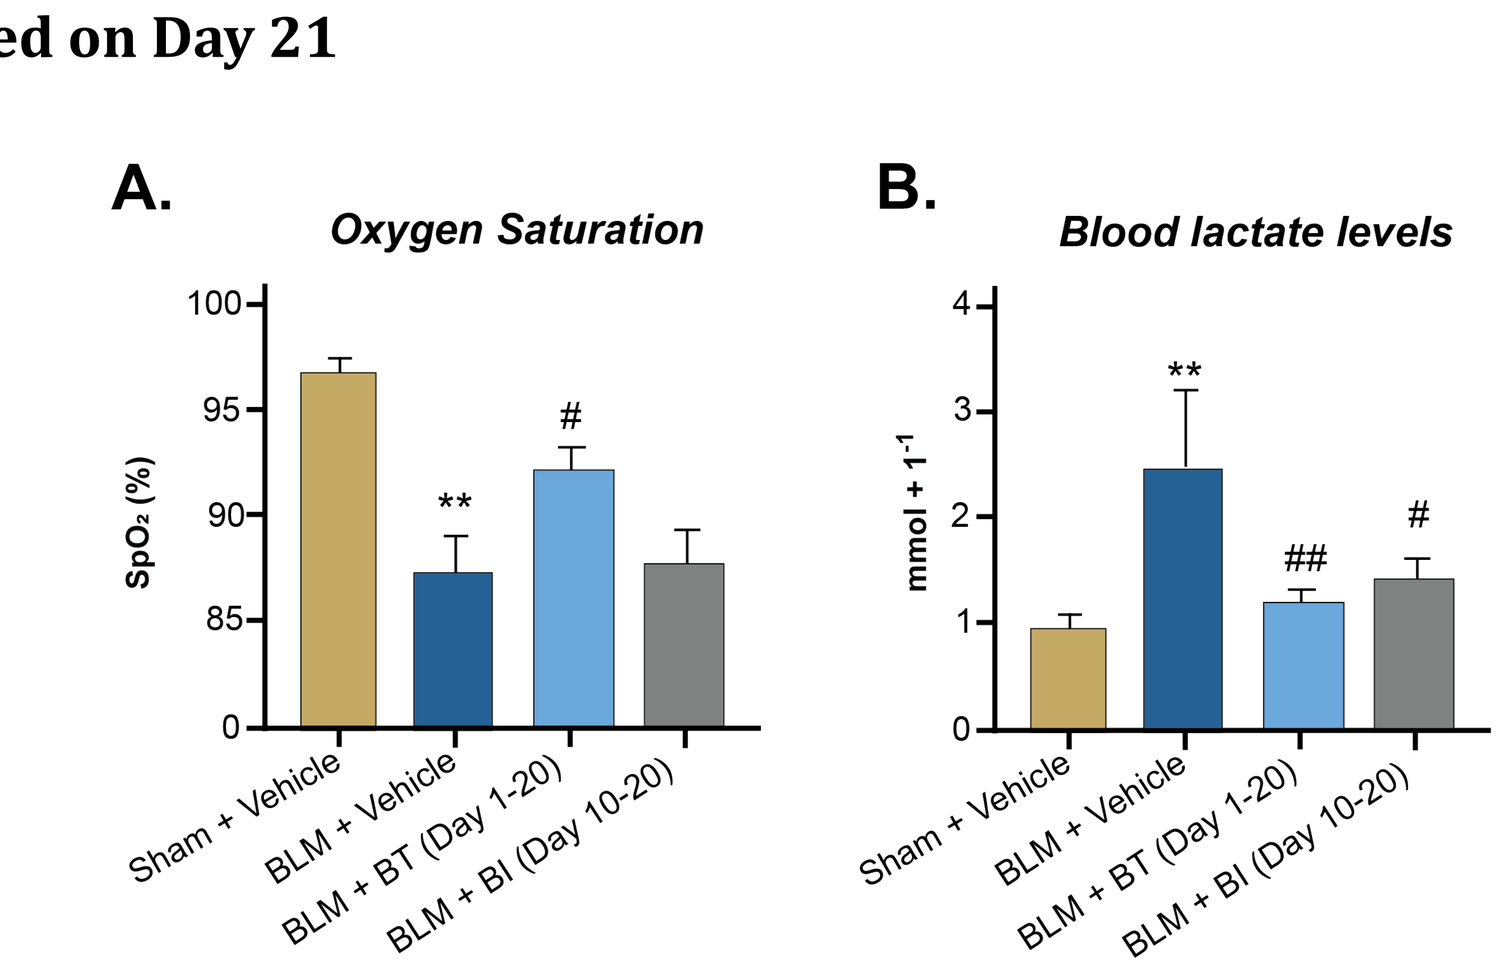
A.
Oxygen Saturation
SpO_2 (%)
B.
Blood lactate levels
mmol + 1-1
Sham + Vehicle
BLM + Vehicle
BLM + BT (Day 1-20)
BLM + BI (Day 10-20)

BLM: Bleomycin; Sham: Non-induced animals with the vehicle.
BT normalized blood oxygen levels (P<0.05, BLM) (Figure 4A).
Both significantly reduced blood lactate levels (BT: P<0.01, BLM; BI: P<0.05, BLM) (Figure 4B).

FIGURE 5

Respiratory resistance (A) lung hydroxyproline (B) measured on Day 21

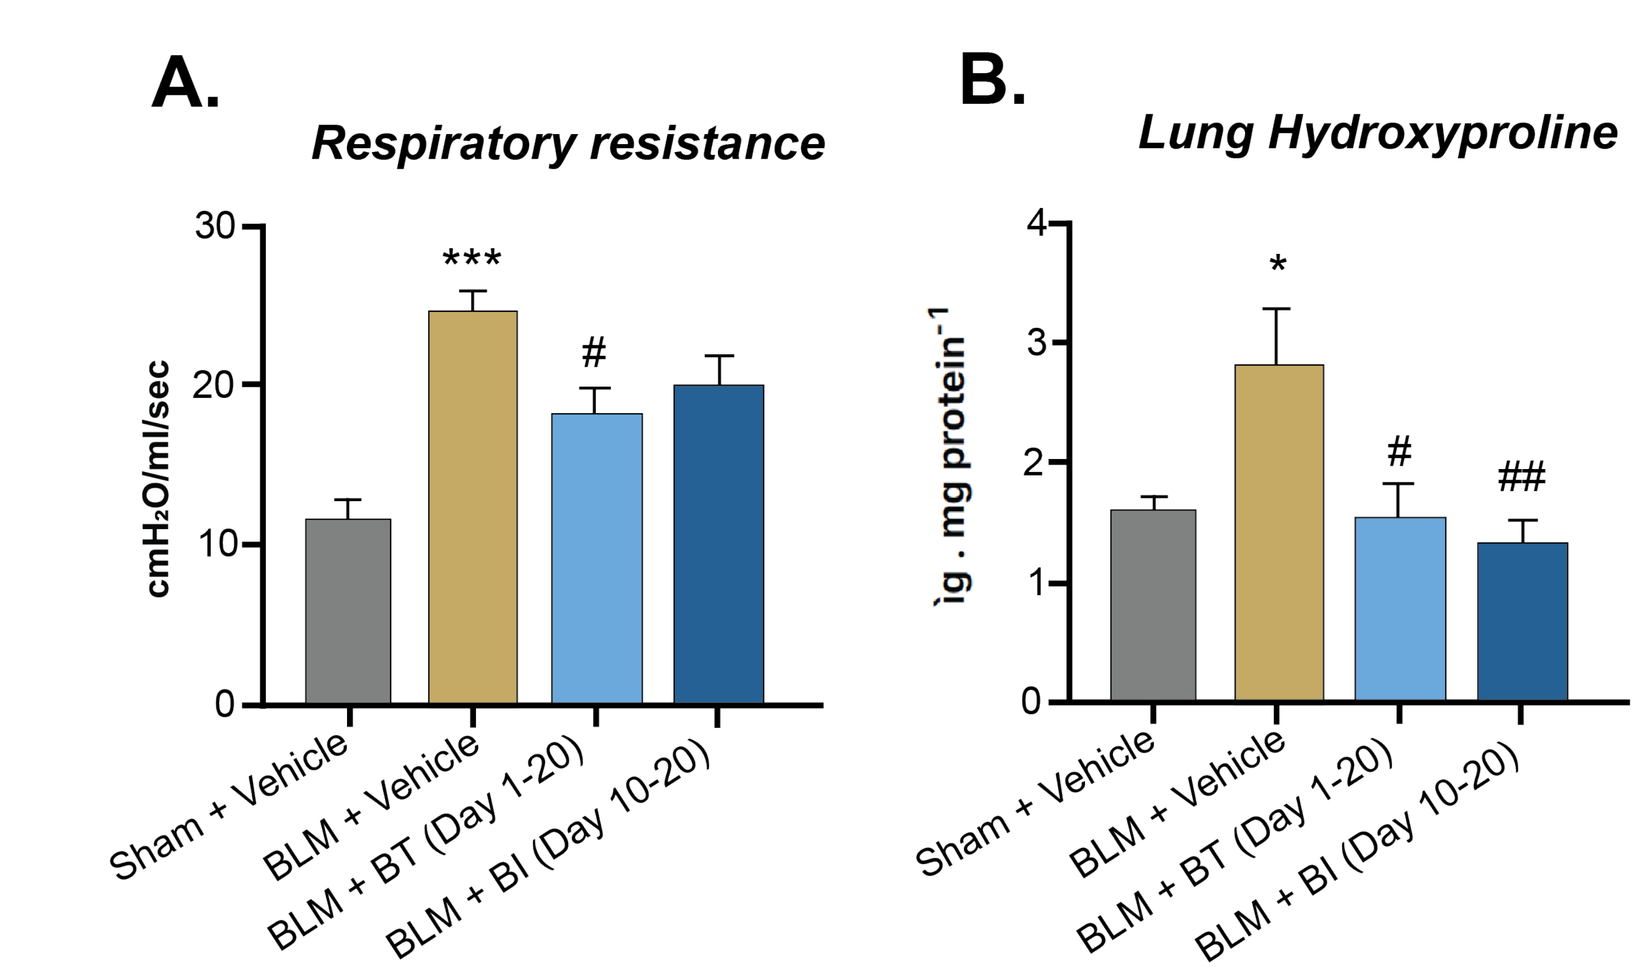
A.
Respiratory resistance
cmH₂O/ml/sec
B.
Lung Hydroxyproline
ìg . mg protein-1
Sham + Vehicle
BLM + Vehicle
BLM + BT (Day 1-20)
BLM + BI (Day 10-20)

BLM: Bleomycin; Sham: Non-induced animals with the vehicle.
*P<0.05 BLM+Veh; as compared to Sham. *** P<0.001; as compared to Sham.
P<0.05, vs. BLM. ## P<0.01, vs. BLM.
BT significantly reduced respiratory resistance (P<0.05, BLM) (Figure 5A). Both (BT: P<0.05, BLM; BI: P<0.01, BLM) significantly decreased hydroxyproline content (Figure 5B).

FIGURE 6

Parameters reflective of pulmonary edema at Day 21 including lung weight (A), BALF cell count (B), and BALF total protein (C)

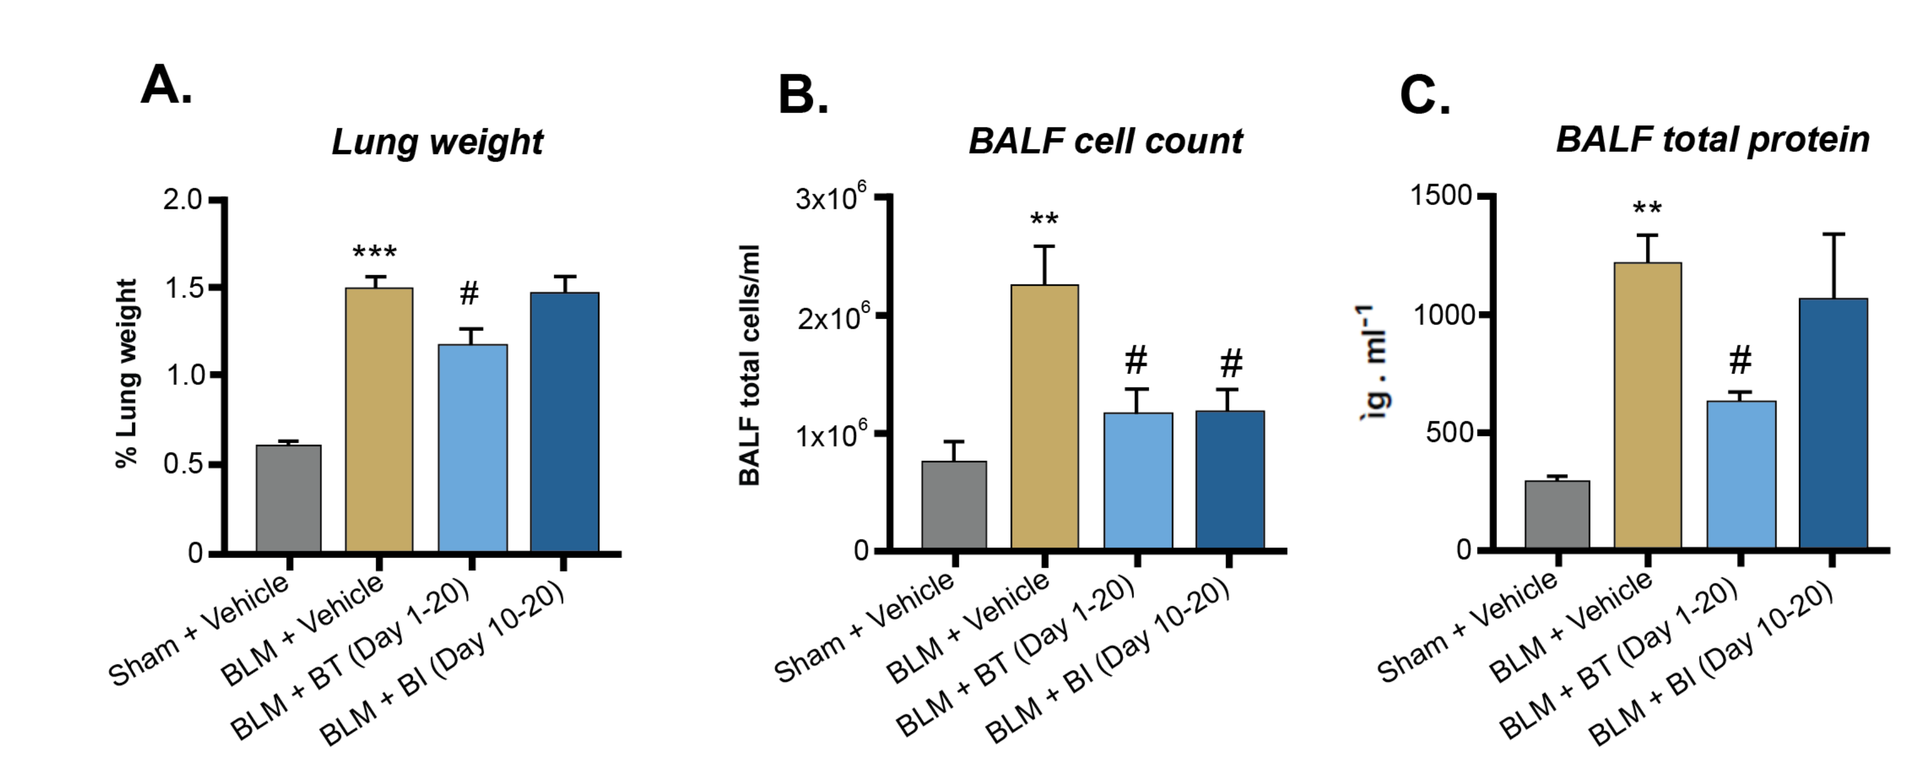
A.
Lung weight
% Lung weight
B.
BALF cell count
BALF total cells/ml
C.
BALF total protein
ìg . ml-1

BALF: Bronchoalveolar lavage; BLM: Bleomycin; Sham: Non-induced animals with the vehicle.
P<0.01, BLM vs. Sham. * P<0.001, vs. Sham. # P<0.05, vs. BLM.
BT lung weight (Figure 6A) was significantly (P<0.05) lower the BLM-induced increase. BT reversed BLM-induced total cell counts and protein levels (Figure 6B, C) (P<0.05, BLM); BI significantly reduced the total cell counts (P<0.05, BLM).

FIGURE 7

Morphology changes displayed by H&E staining and Ashcroft Score (A, C) and collagen deposition and Masson's trichrome staining (B, D) induced by BLM in rats on Day 21

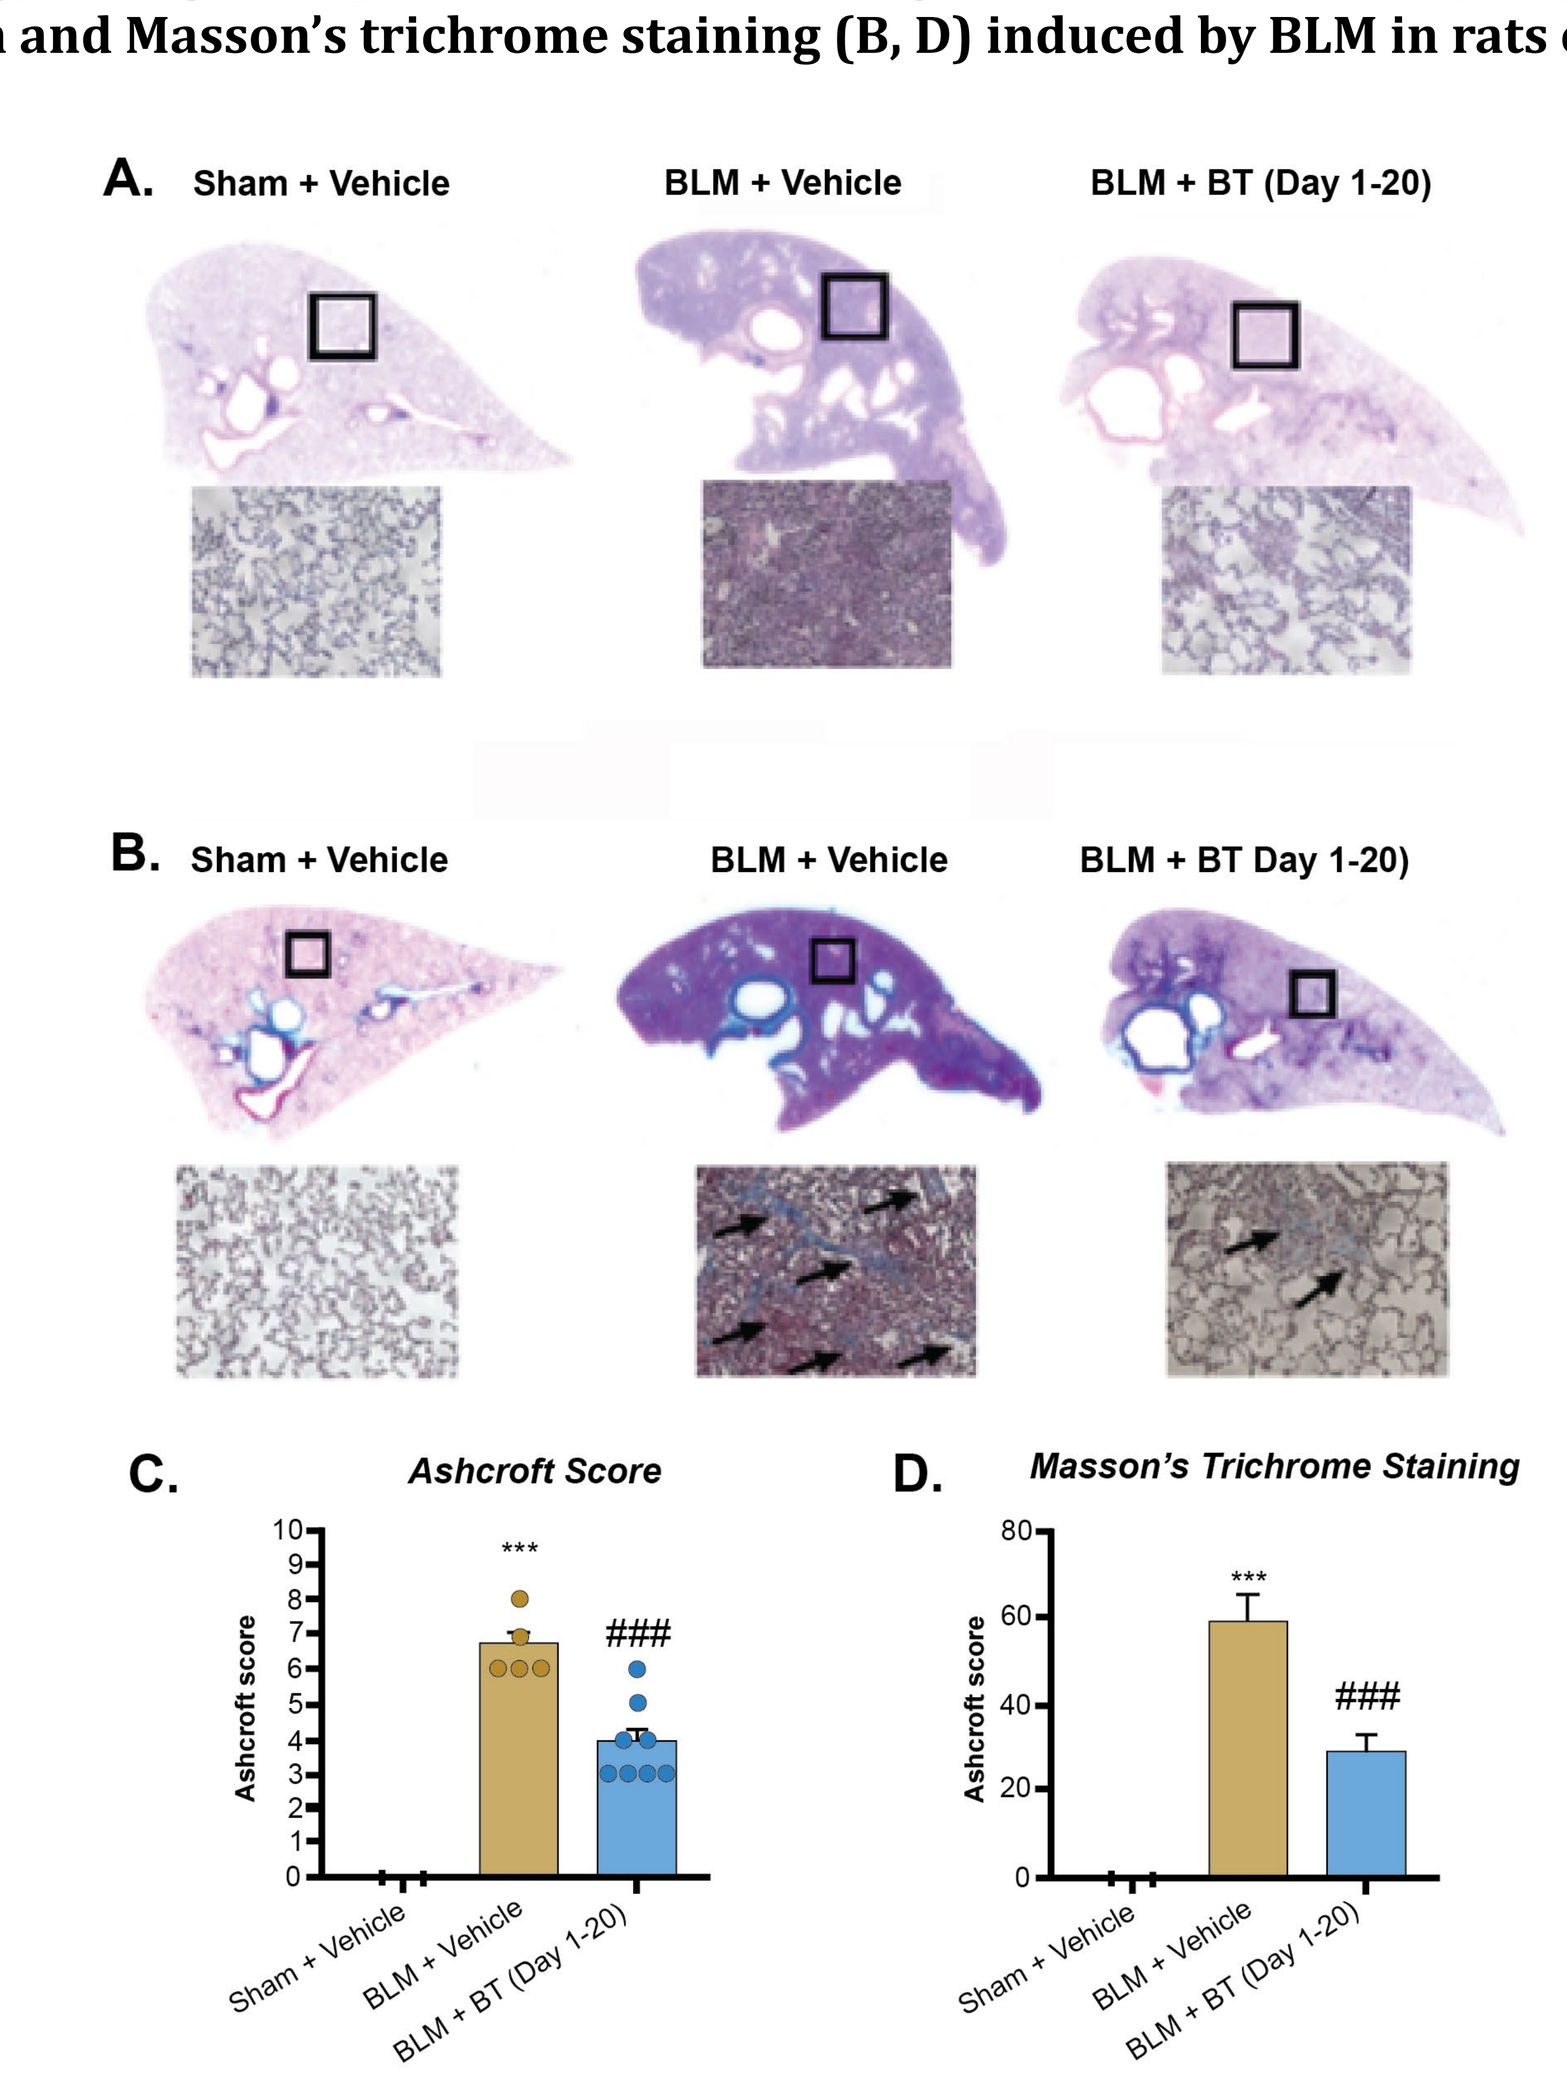
A. Sham + Vehicle
BLM + Vehicle
BLM + BT (Day 1-20)
B. Sham + Vehicle
BLM + Vehicle
BLM + BT Day 1-20)
C.
Ashcroft Score
D.
Masson's Trichrome Staining

BLM: Bleomycin; Sham: Non-induced animals with the vehicle.

*** P<0.001, vs. Sham. ### P<0.001, vs. BLM.

BT significantly (P<0.001) reduced BLM-induced fibrotic changes based on Ashcroft Score and Masson's trichrome staining.

FIGURE 8

BALF cytokine levels on Day 21: MIP1 (A); MCP1 (B); IL6 (C); IP10 (D); and RANTES (E)

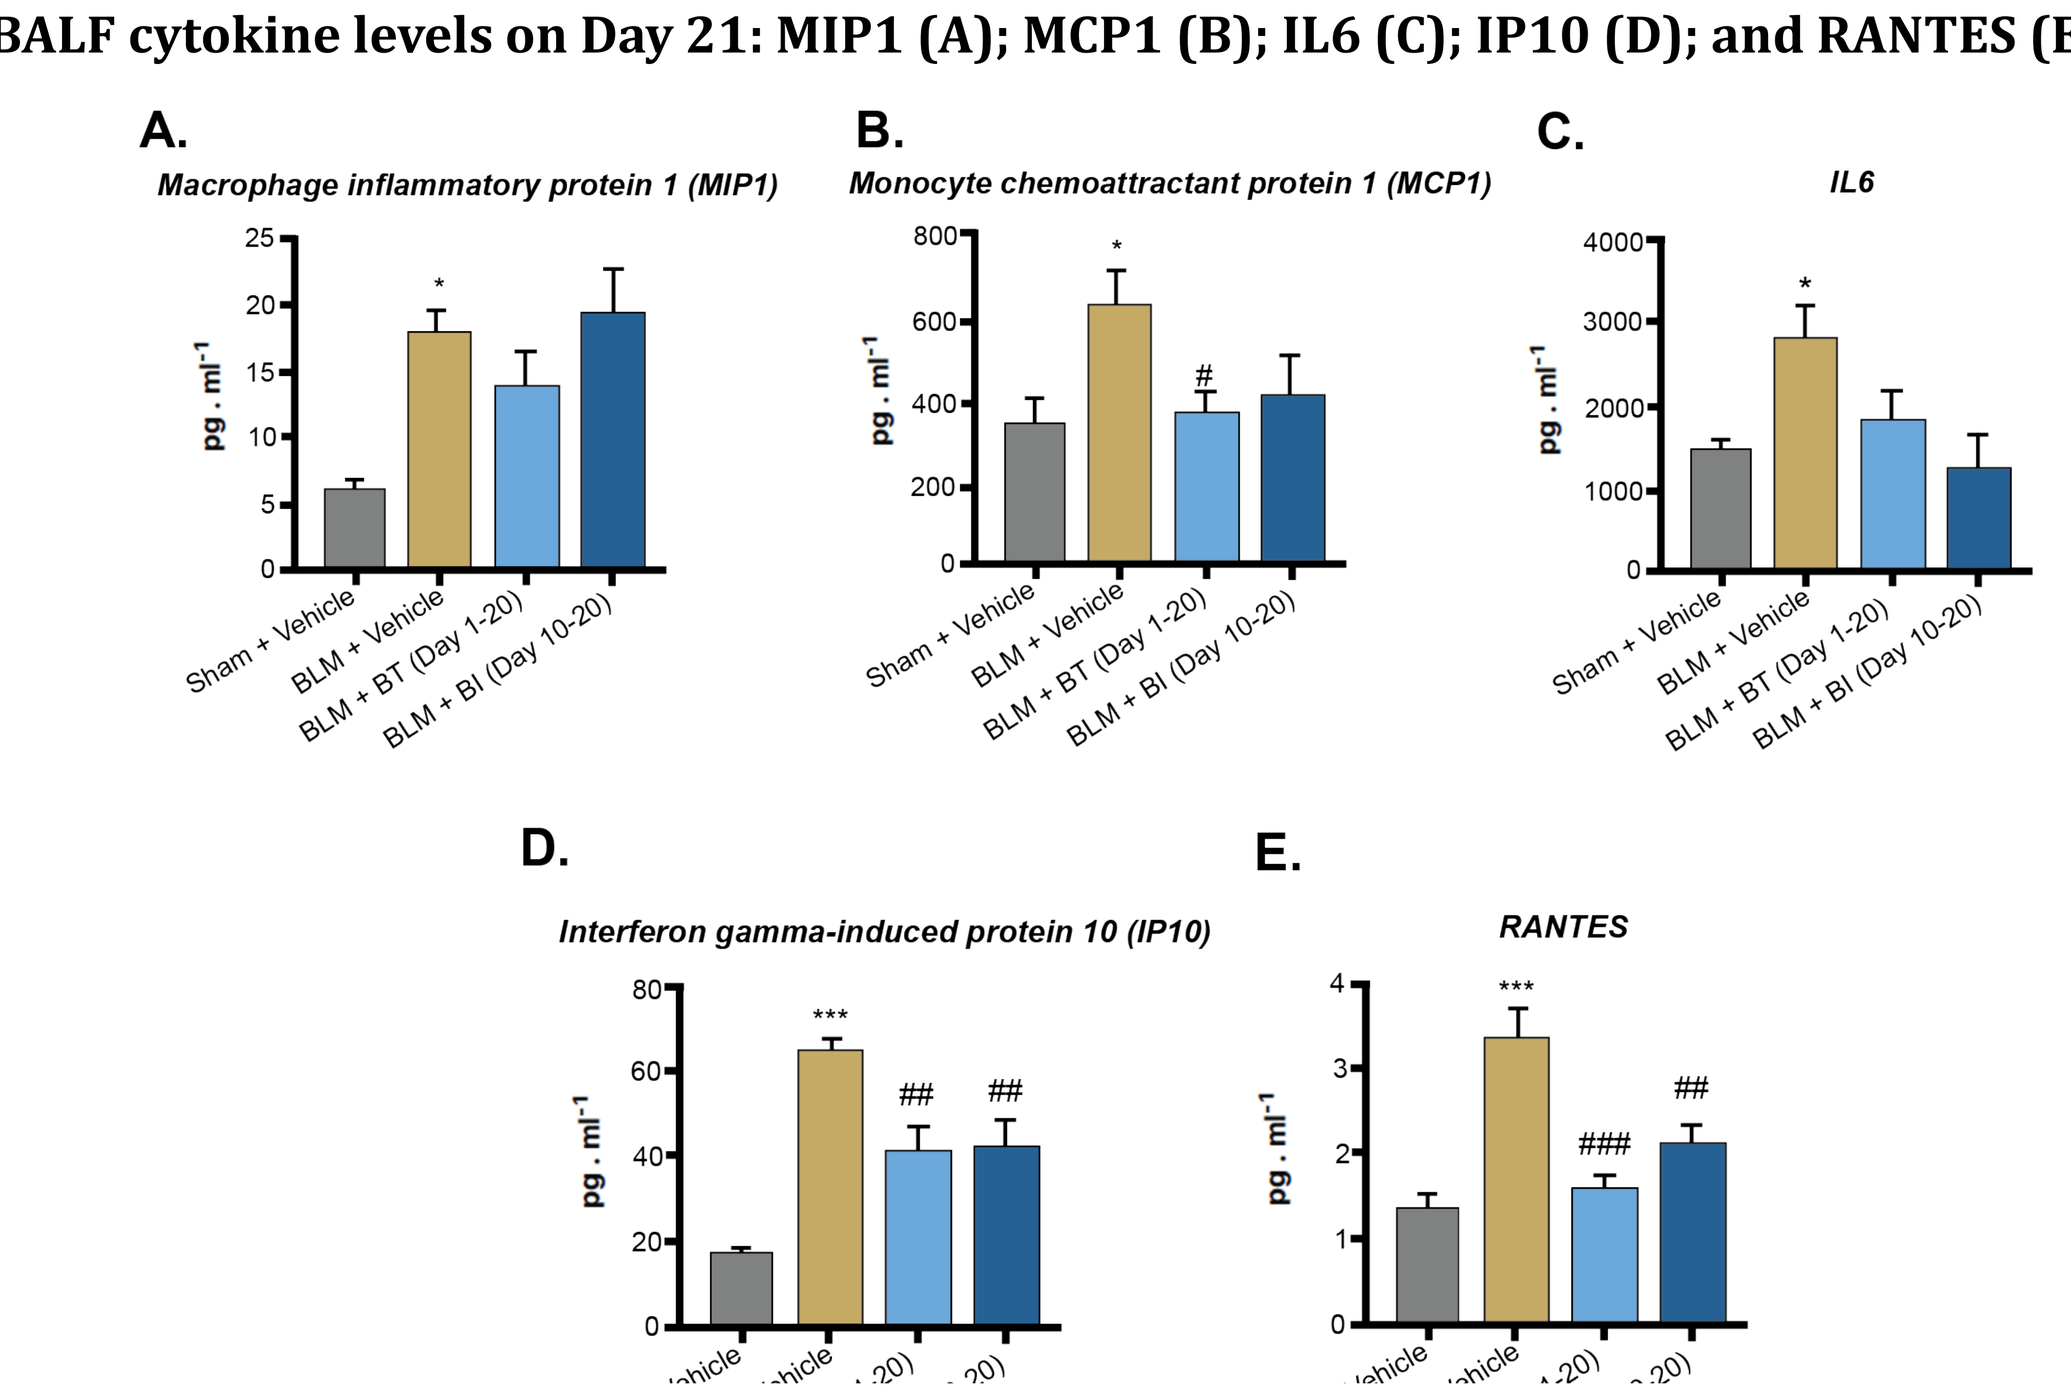
A.
Macrophage inflammatory protein 1 (MIP1)
B.
Monocyte chemoattractant protein 1 (MCP1)
C.
IL6
D.
Interferon gamma-induced protein 10 (IP10)
E.
RANTES
pg . ml-1

BLM: Bleomycin; Sham: Non-induced animals with the vehicle.
* P<0.05, vs. Sham. *** P<0.001, vs. Sham.
P<0.05, vs. BLM. ## P<0.01, vs. BLM. ### P<0.001, vs. BLM
BT significantly decreased MCP-1 (P<0.05, BLM) (Figure 8B). Both BT and BI significantly reduced (IP10 and RANTES P<0.01, BLM) (Figure 8D, E)

DISCUSSION

This study was the first preclinical evaluation of brilaroxazine's effects on IPF and was consistent with standard models[19,20].

Brilaroxazine impacted mortality and functionality, plus inflammation and fibrosis. It positively affected survival, body weight, lung edema, fibrogenic cytokine production, hydroxyproline content, respiratory resistance, and cardiopulmonary capacity. BT effects were significant across a broader parameter array; BI's impact may be limited due to lower BLM animal numbers (due to mortality) and Day 21 sacrifice.

Improvements might be related to the multiple effects via 5-$HT_{2B/7}$[9,21,22], which other 5-HT_{2B} receptor antagonists have shown[22,23]. The decrease in BLM-induced up-regulated hydroxyproline content support brilaroxazine's antifibrotic role. Reduction of multiple cytokines in this study (IL-6, INF-γ [IP10], MIP1, MCP1, RANTES) and brilaroxazine's other works in pulmonary arterial hypertension and psoriasis, supports its anti-inflammatory action[16-18].

5-HT signaling pathology leads to broncho- and pulmonary arterial constriction, myofibroblast, and smooth muscle cell hypertrophic and hyperplastic alterations[9,22,25]. These effects involve myofibroblast inflammation, proliferation, fibrogenesis involving extracellular matrix deposition, and pulmonary vascular endothelial and smooth muscle cell effects.

CONCLUSION

Brilaroxazine, via its actions on fibrosis and inflammation involving the pulmonary vasculature and myofibroblasts via 5-$HT_{2B/7}$, significantly improved key endpoints and biomarkers in this BLM-induced IPF.

REFERENCES

1. Moore BB et al. (2013). Am J Respir Cell Mol Biol. 49(2):167-179.
2. Bors M et al. (2015). Chron Respir Dis. 12(4):365-372.
3. Hutchinson J et al. (2015). European Respiratory Journal. 46(3):795-806.
4. Nalysnyk L et al. (2012). European Respiratory Review. 21(126):355-361.
5. Puglisi S et al. (2016). Ther Adv Chronic Dis. 7(2):108-120.
6. Raghu G et al. (2006). Am J Respir Crit Care Med. 174(7):810-816.
7. Meltzer EB et al. (2008). Orphanet J Rare Dis. 3(1):1-15.
8. Sakai N et al. (2013). Biochimica et Biophysica Acta (BBA)-Molecular Basis of Disease. 1832(7):911-921.
9. Mann DA et al. (2013). Biochimica et Biophysica Acta (BBA)-Molecular Basis of Disease. 1832(7):905-910.
10. Dees C et al. (2011). Journal of Experimental Medicine. 208(5):961-972.
11. Königshoff M et al. (2010). Thorax. 65(11):949-955.
12. Pan J et al. (2006). American Journal of Physiology-Lung Cellular and Molecular Physiology. 290(1):L185-L193.
13. Cantillon M et al. (2017). Schizophr Res. 189:126-133.
14. Köster LS et al. (2014). Expert Opin Emerg Drugs. 19(4):511-531.
15. Bhat L et al. (2017a). Eur J Pharmacol. 810:92-99.
16. Bhat L et al. (2017b). Eur J Pharmacol. 810:83-91.
17. Bhat L et al. (2018). Eur J Pharmacol. 827:159-166.
18. Bhat L et al. (2023). International Society of Investigative Dermatology meeting, poster#LB1745.
19. Moeller A et al. (2008). Int J Biochem Cell Biol. 40(3):362-382.
20. Liu Y et al. (2017). BMC Pulm Med. 17:1-11.
21. Tzouvelekis A et al. (2015). Ther Clin Risk Manag. e359-e370.
22. Wynn TA. (2011). Journal of Experimental Medicine. 208(7):1339-1350.
23. Dumitrascu R et al. (2011). European Respiratory Journal. 37(5):1104-1118.
24. Fabre A et al. (2008). European Respiratory Journal. 32(2):426-436.
25. Löfdahl A et al. (2016). Physiol Rep. 4(15):e12873.

DISCLOSURES & ACKNOWLEDGEMENTS

Conflict of Interest: Laxminarayan Bhat and Seema R Bhat are employees of Reviva Pharmaceutical Holdings, Inc.

Funding Source: This study's grant was awarded to ISPT from Reviva Pharmaceutical Holdings, Inc.

Acknowledgments: John M. York, PharmD, MBA, Akita Biomedical, Inc. provided editorial support, and Reviva Pharmaceutical Holdings, Inc. funded this effort.