Absorption, Metabolism, and Excretion of Brilaroxazine in Rats

Laxminarayan Bhat*, Seema Bhat, and Palaniappan Kulanthaivel

Reviva Pharmaceuticals, Inc., 10080 N Wolfe Road, Suite SW3-200, Cupertino, California, USA

bit.ly/ASPETPOSTERPMME

ASPET Poster # 491 | May 18, 2024
ADDC Poster # 807 | May 19, 2024
Abstract ID: 129061

BACKGROUND

Brilaroxazine, a novel multimodal neuromodulator, is part of the next-generation treatments for schizophrenia and comorbid conditions.[1-2] This novel multimodal serotonin (5-HT) and dopamine (D) receptor modulator mitigates multiple inflammatory cytokines.[1-9] It specifically functions as a $D_{2/3/4}$ and 5-$HT_{1A/2A}$ partial agonist and a 5-$HT_{2B/7}$ antagonist with binding affinity for 5-HT_{2B} > D_2.[1-4]

This agent possesses an established efficacy, safety, and pharmacokinetic (PK) profile based on its Phase 1 and 2 clinical experience, including the Phase 2 REFRESH study.[1,2,10] It possesses differentiated pharmacological and safety profiles over other antipsychotics.[1-10] The Phase 3 RECOVER study indicates 50 mg brilaroxazine daily is highly effective and well tolerated and offers neuroinflammatory capabilities.[8,9]

Brilaroxazine's nonclinical toxicological evaluations involved rats, mice, and dogs as animal models. Single-dose comparison studies between animals (mice and dogs) and humans, provide insight into brilaroxazine's PK, mass balance, metabolism, and excretion profiles.[11] In these studies, feces represent the predominant route of excretion, with M465a as the major excreted metabolite.[11] M219 is the major circulating metabolite in all species, and no human-specific metabolite appears to exist in plasma.[11] The emergent metabolic pathways for all species involve oxidation, N- or O-dealkylation with subsequent sulfation, and/or conjugation with glucuronic acid.[11] Such data qualified mice and dogs for preclinical work. Still, the need for similar data in rats exists to qualify this animal group as a suitable toxicology species.

OBJECTIVE

This research evaluated brilaroxazine's mass balance, metabolism, and excretion profile in intact and bile duct-cannulated (BDC) male Sprague Dawley rats after a single ^{14}C-brilaroxazine dose.

METHODS

Regulatory

Animal studies occurred at an Association for Assessment and Accreditation of Laboratory Animal Care facility and in compliance with Institutional Animal Care and Use Committees protocols. The human study had approval from the investigational review board. The study ran under the guidance of the US Nuclear Regulatory Commission and the State Bureau of Radiation Protection regulations (Pennsylvania).

Procedures

The study involved 12 adult male Sprague Dawley (SD) rats (Charles River, Raleigh, NC) divided into three separate groups to evaluate a single oral 20 mg/kg (~200 µCi/kg) dose of [^{14}C]brilaroxazine.

- Group 1 animals had BDC surgically implanted seven days prior to [^{14}C]brilaroxazine dosing. After the radioactive dose, they received bile salts (44 mM of cholic acid in saline, pH ~ 7.6) as replenishment during the study. Investigators collected bile for up to 72 hours, urine, feces, and/or cage rinse at various time intervals up to 168 hours.

 Bile: pre-dose, 0-4, 4-8, 8-24, 24-48, & 48-72 hr.
 Urine: pre-dose, 0-8, 8-24 and daily up to 168 hr.
 Feces: pre-dose, 0-24, and daily up to 168 hr.
 Cage Rinse: at the end of 168 hr.

- Group 2 had femoral artery cannulas (FAC) implanted for at least two days prior to [^{14}C]brilaroxazine dosing. Investigators collected whole blood in tubes containing K2EDTA anticoagulant at each time point (N=4) up to 72 hr, pooled at each time point, and then centrifuged to obtain plasma.

 Blood/plasma: pre-dose, 0.25, 0.5, 1, 2, 4, 8, 12, 24, 48, and 72 hr. (N=4/time point)

- Group 3 consisted of intact animals with no surgical implantations prior to [^{14}C]brilaroxazine dosing. Investigators collected urine, feces, and/or cage rinse at various time intervals up to 168 hr.

 Urine: pre-dose, 0-8, 8-24, and daily up to 168 hr.
 Feces: pre-dose, 0-24 and daily up to 168 hr.
 Cage Rinse: at the end of 168 hr.

Analysis

Determination of radioactivity concentrations of [^{14}C]brilaroxazine and its metabolites from blood, plasma, and excreta utilized liquid scintillation counting (LSC) or a combination of combustion with LSC. Plasma concentration assessment of unlabeled brilaroxazine and its metabolite (RP5081) used a validated liquid chromatography/tandem mass spectrometry method. Identification of brilaroxazine-related metabolite structures in biological samples involved mass spectral analysis or via direct comparison with authentic standards for metabolite structures (brilaroxazine and metabolites RP5081, M219 and M465a). Plasma PK parameter estimates of total radioactivity (TRA) and brilaroxazine employed non-compartmental analysis (WinNonlin Software, Certara, Princeton, NJ).

RESULTS

Figure 1: Mass Balance

Figure 1. Cumulative recovery of TRA after a single oral dose of 20 mg/kg [^{14}C]brilaroxazine in male (a) BDC and (b) intact SD rats.

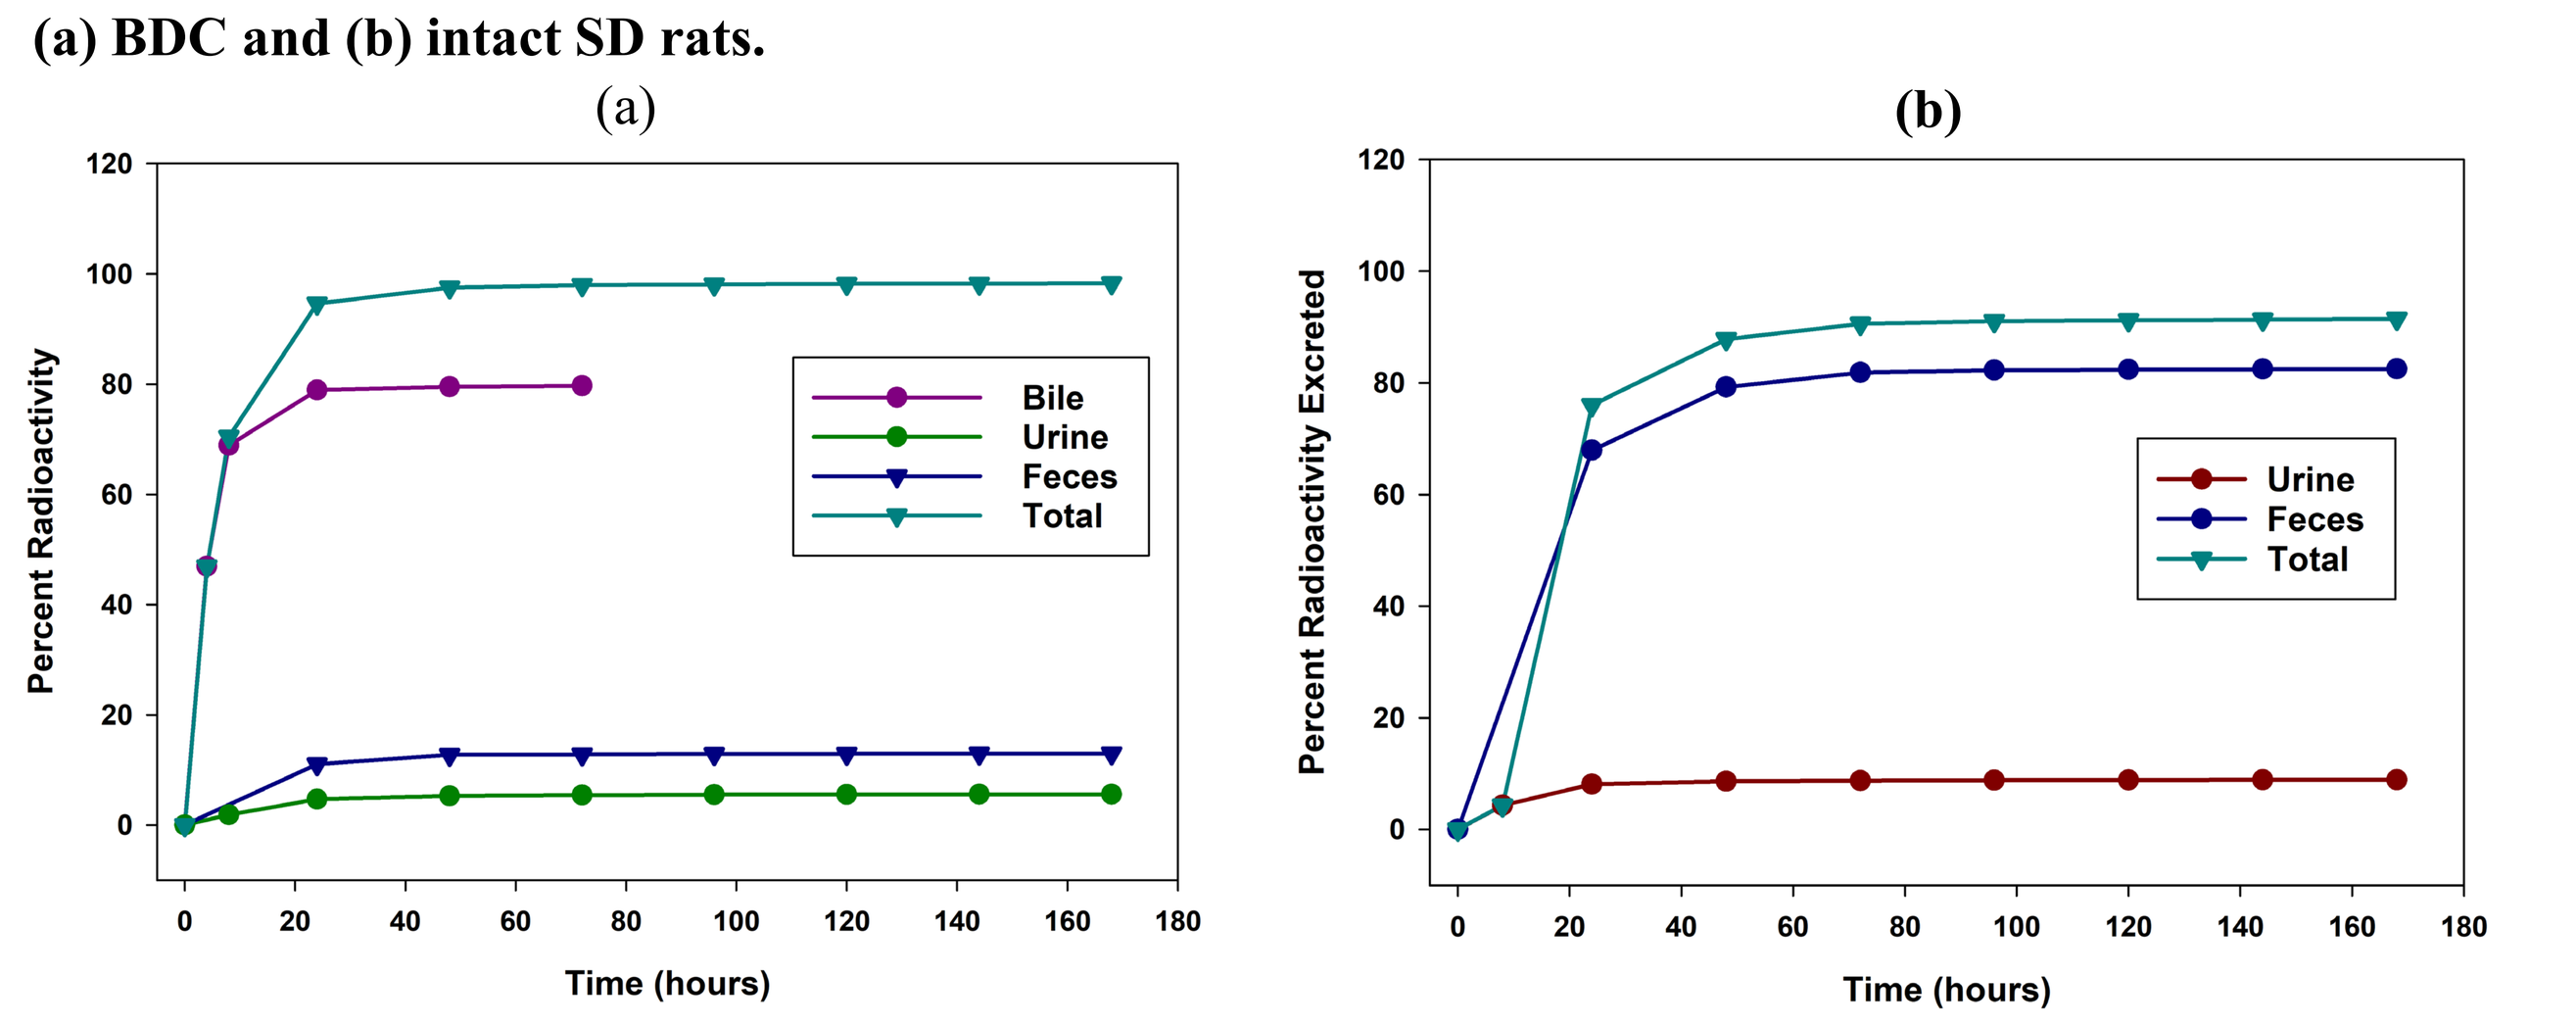
(a)
(b)
Percent Radioactivity
Percent Radioactivity Excreted
Time (hours)
Bile
Urine
Feces
Total

Bile or feces of BDC rats or intact rats was the predominant route for radioactivity excretion. The percent of the administered dose recovered in Group 1 BDC rat bile, feces, and urine accounted for 79.52%, 13.02%, and 5.60%, respectively (total recovered 98.33%). The administered dose recovered in Group 3 intact rat feces and urine accounted for 81.51% and 8.89%, respectively (total recovered 91.48%). The amount of dose recovered in bile and urine in BDC rats indicate that at least 85% of administered dose was absorbed.

Figure 2: Pharmacokinetics

Figure 2. Plasma concentration-time profile of TRA, brilaroxazine and metabolite RP5081 following a single oral dose of 20 mg/kg [^{14}C]brilaroxazine in male intact SD rats.

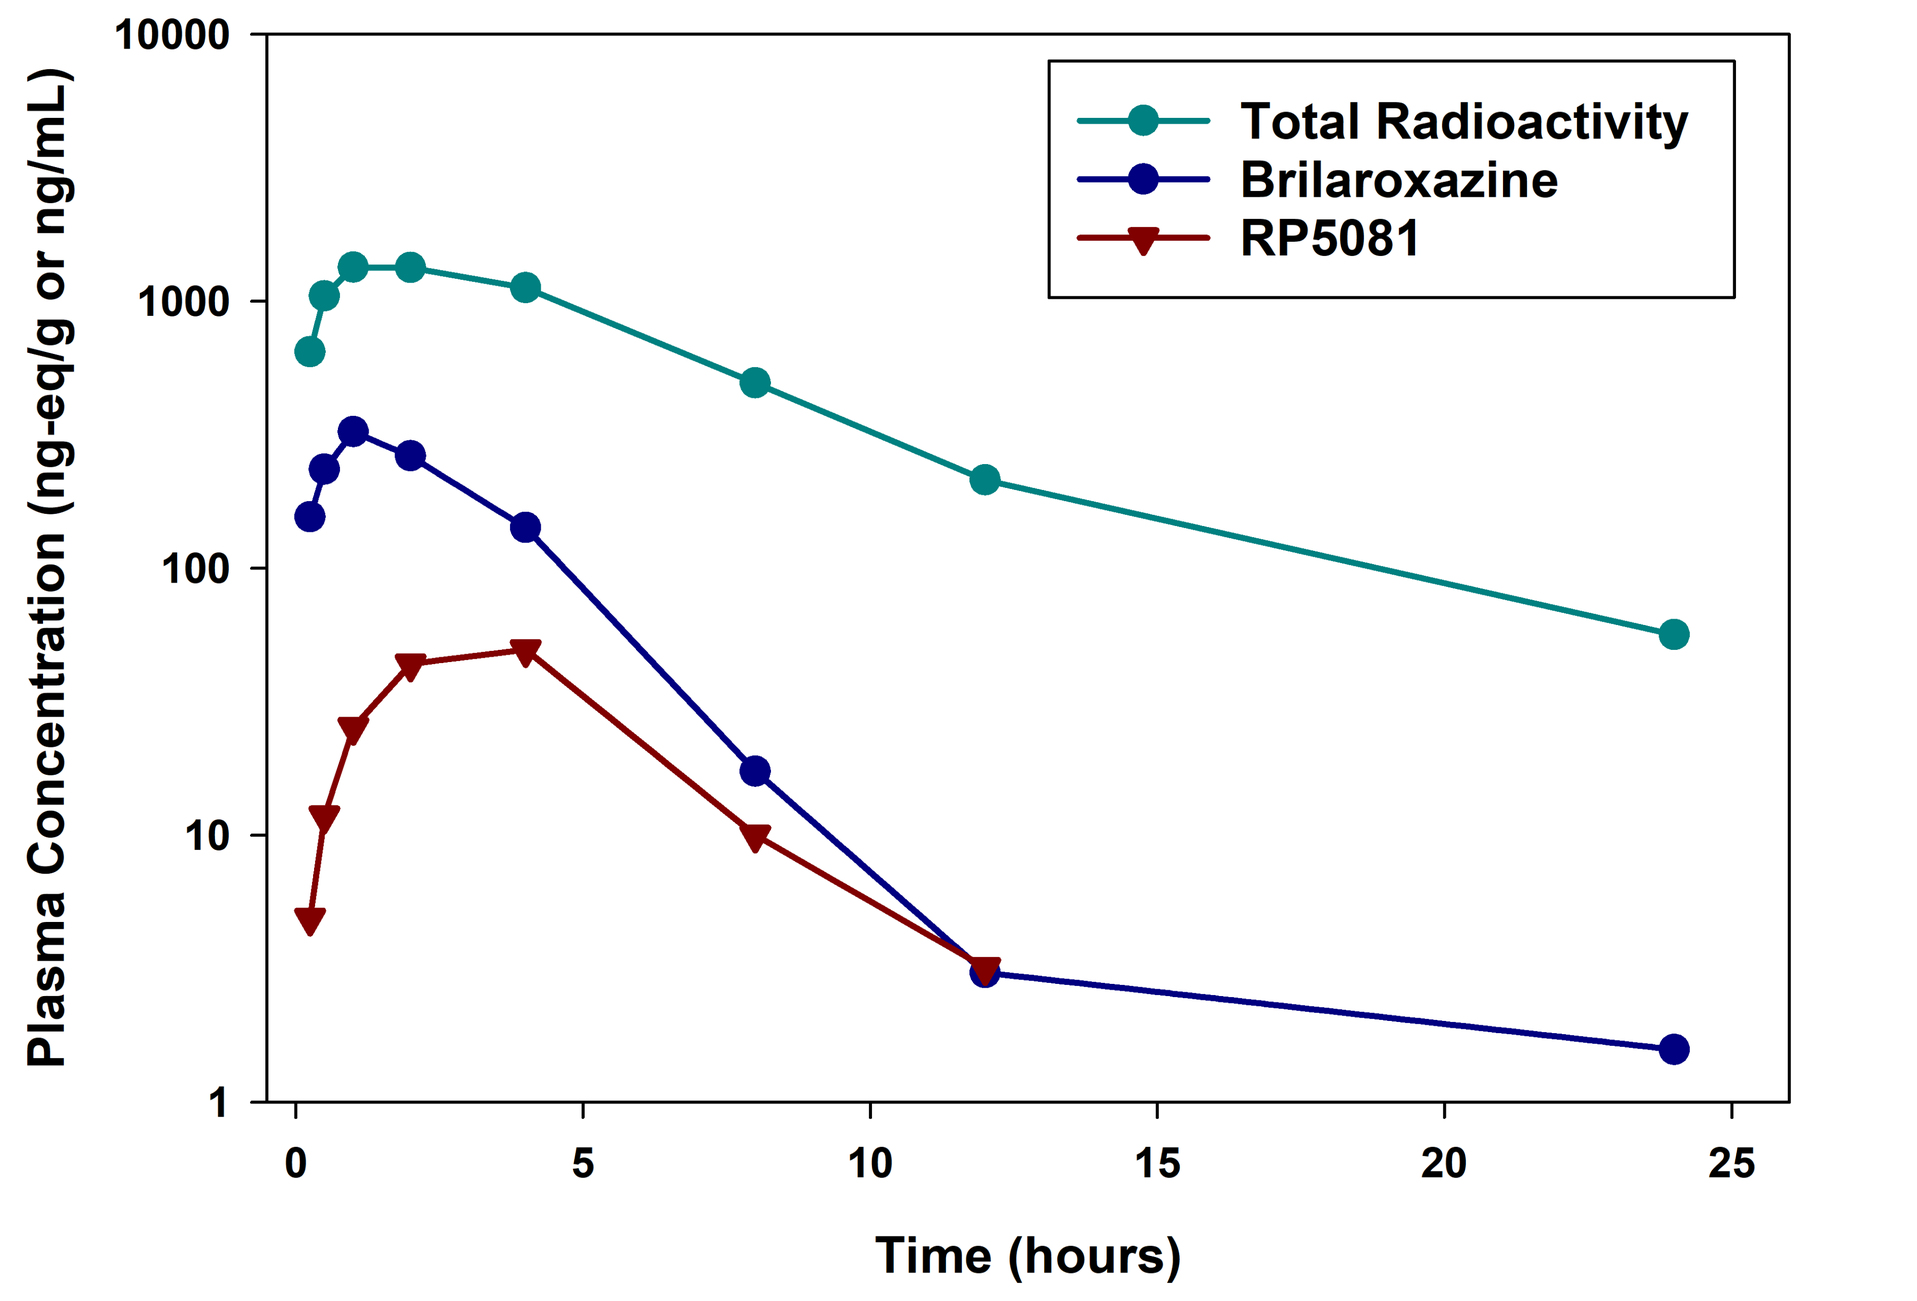
Plasma Concentration (ng-eq/g or ng/mL)
Time (hours)
Total Radioactivity
Brilaroxazine
RP5081

Figure 3: Metabolism

Figure 3. Radio-chromatogram of pooled plasma samples ($AUC_{0\text{-}24hr}$) from male SD rats following a single oral dose of [^{14}C]brilaroxazine at 20 mg/kg (~200 µci/kg)

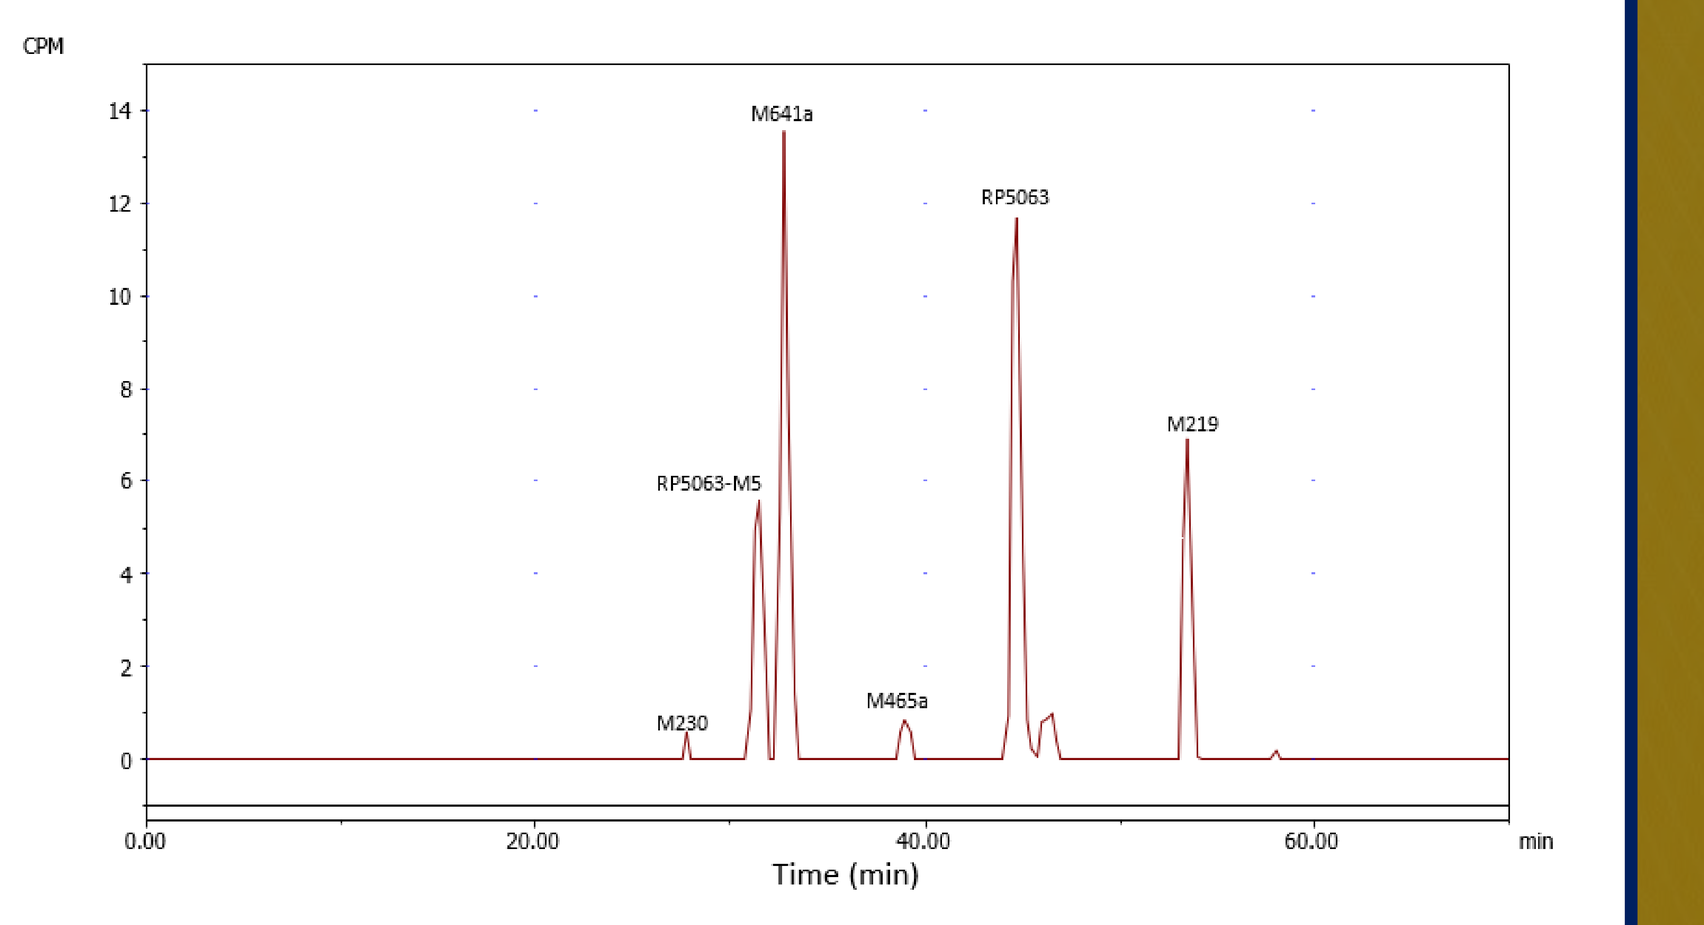
CPM
M641a
RP5063
RP5063-M5
M219
M230
M465a
Time (min)

Brilaroxazine represents ~31% of the circulating radioactivity exposure, with three primary metabolites: M641a (oxidation + glucuronidation), RP5081 (RP5063-M5; O-dealkylated acid), and M219 (N-[2,3-dichlorphenyl]-glycine) accounting for ~29%,~16%, and ~16%, respectively.

Figure 4: Metabolism

Figure 4. Radio-chromatogram of pooled bile samples (representing >90% of biliary radioactivity) from BDC Rats following a single oral dose of [^{14}C]brilaroxazine at 20 mg/kg (~200 µci/kg)

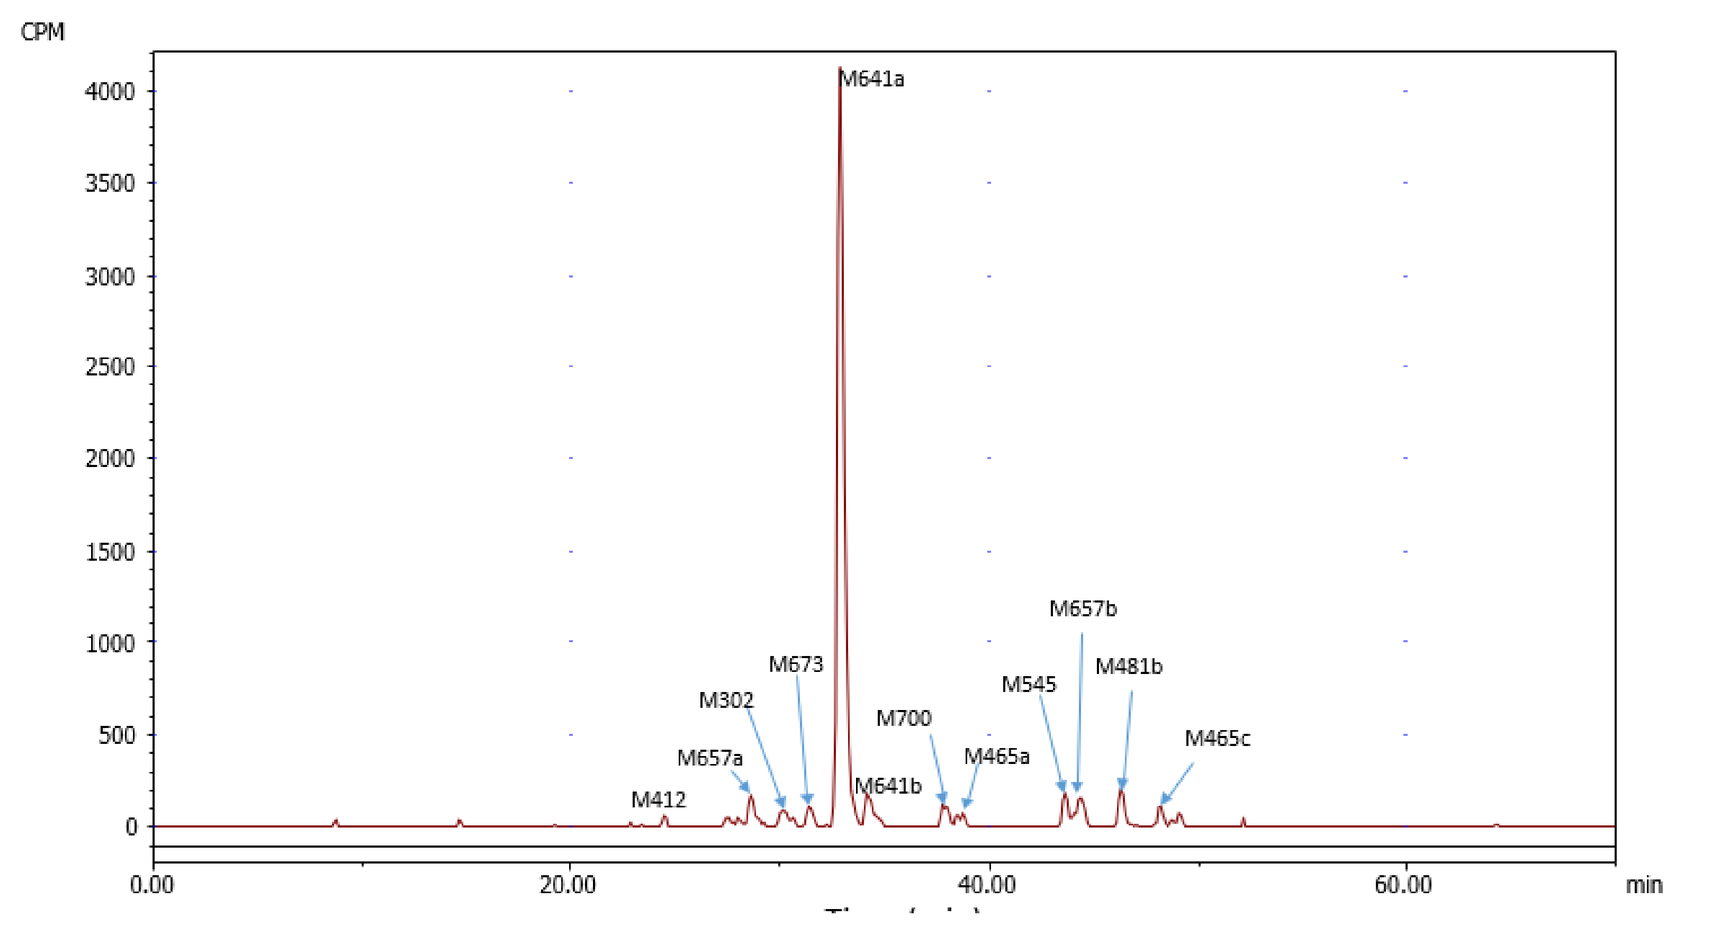
CPM
M641a
M657b
M481b
M673
M302
M700
M545
M657a
M465a
M465c
M412
M641b

The analysis failed to detect brilaroxazine but identified mono-hydroxylated glucuronide metabolite, M641a, as the major component accounting for 70% of the bile radioactivity, or 56% of the dose. Also detected were several minor metabolites accounting for <3% of the dose.

Figure 5: Metabolism

Figure 5 . Radio-chromatogram of pooled urine samples (representing >90% of urine radioactivity) from intact rats (b) following a single oral dose of [^{14}C]brilaroxazine at 20 mg/kg (~200 µci/kg)

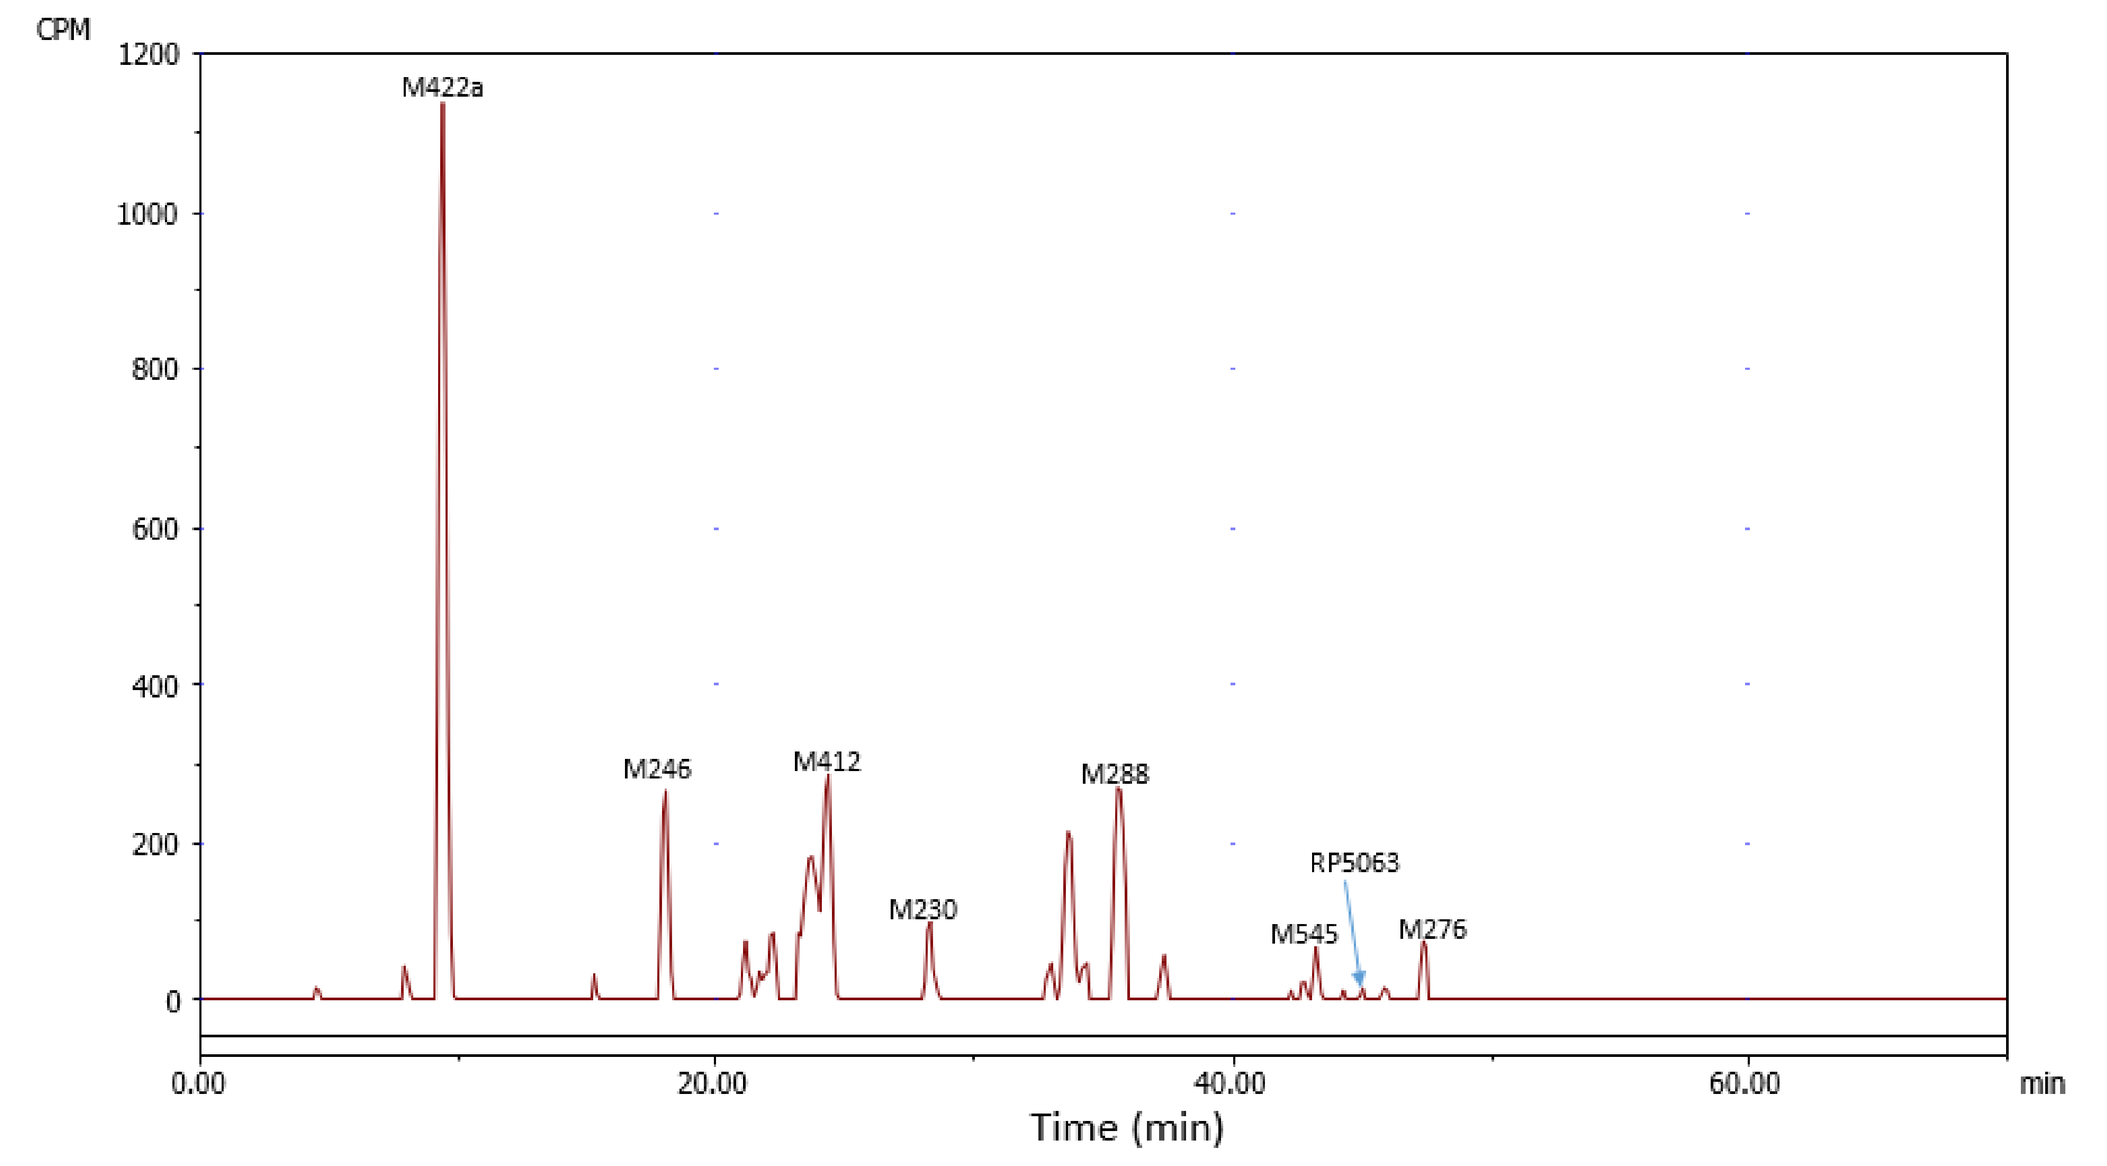
CPM
M422a
M246
M412
M288
M230
RP5063
M545
M276
Time (min)

Brilaroxazine's excretion was at trace levels (<0.3% of the dose) in rat urine. Multiple metabolites, each accounting for <1% of the dose, were also excreted in urine.

Figure 6: Metabolism

Figure 6 . Radio-chromatogram of pooled fecal samples (representing >90% of fecal radioactivity) from intact rats following a single oral dose of [^{14}C]brilaroxazine at 20 mg/kg (~200 µci/kg)

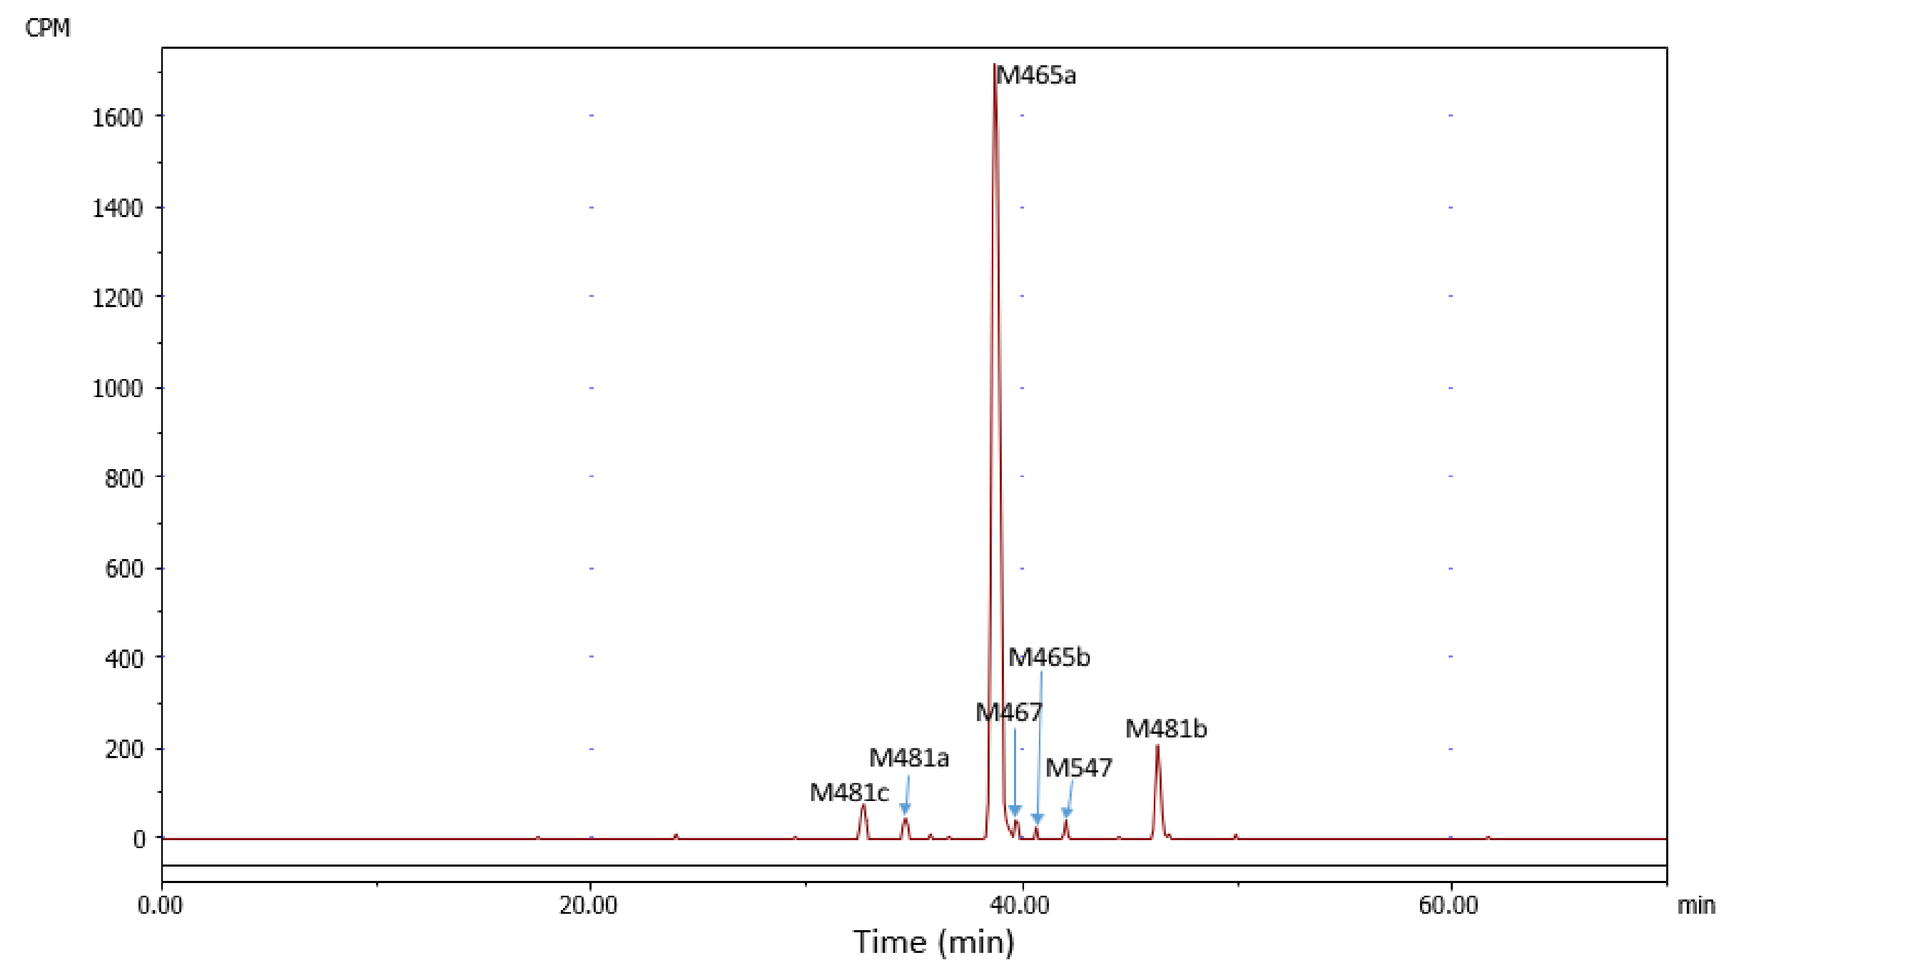
CPM
M465a
M465b
M467
M481b
M481a
M481c
M547
Time (min)

The investigation did not detect brilaroxazine in rat feces. The mono-hydroxylated metabolite, M465a, was the major metabolite, accounting for 86% of the fecal radioactivity or 71% of the dose. Six additional metabolites (M465b, M467, M481a-c and M547), each accounting <5% of the dose, were also detected in rat feces.

Table 1: Pharmacokinetics

Table 1. Summary PK profile of TRA, brilaroxazine and RP5081 in plasma of male rat following a single oral [^{14}C]brilaroxazine 20 mg/kg dose

Compound	t1/2 (hr)	tmax (hr)	tlast (hr)	Cmax (ng Eq/g)	AUClast (h*ng Eq/g)	AUCInf (hr*ng Eq/g)	AUC Extra (%)
Total Radioactivity	5.3	1.00	24.0	1340	11000	11400	**4**
Brilaroxazine	5.41*	1.00	24.0	325	1300	1310	**1**
RP5081	**NC**	**4.00**	**12.0**	**49.5**	**285**	**NC**	**NC**

**Individuals should view this value cautiously since the adjusted R squared was less than 0.8. NC: Not calculated since there was insufficient data to define the elimination phase.*

Figure 2 displays the concentration-time profiles of TRA, brilaroxazine and metabolite RP5081. Table 1 highlights the corresponding key PK parameters. TRA and brilaroxazine reached t_{max} at 1 hr and RP5081 later at 4 hr. Brilaroxazine exposure (AUC_{last}) represents ~12% of TRA exposure, indicating that most of the circulating radioactivity is due to metabolites. Metabolite RP5081 represents ~3% of the TRA exposure (AUC_{last}).

Figure 7

Figure 7. Proposed metabolic pathways of [^{14}C] brilaroxazine in male SD rats following a single oral dose at 20 mg/kg (~200 µci/kg)

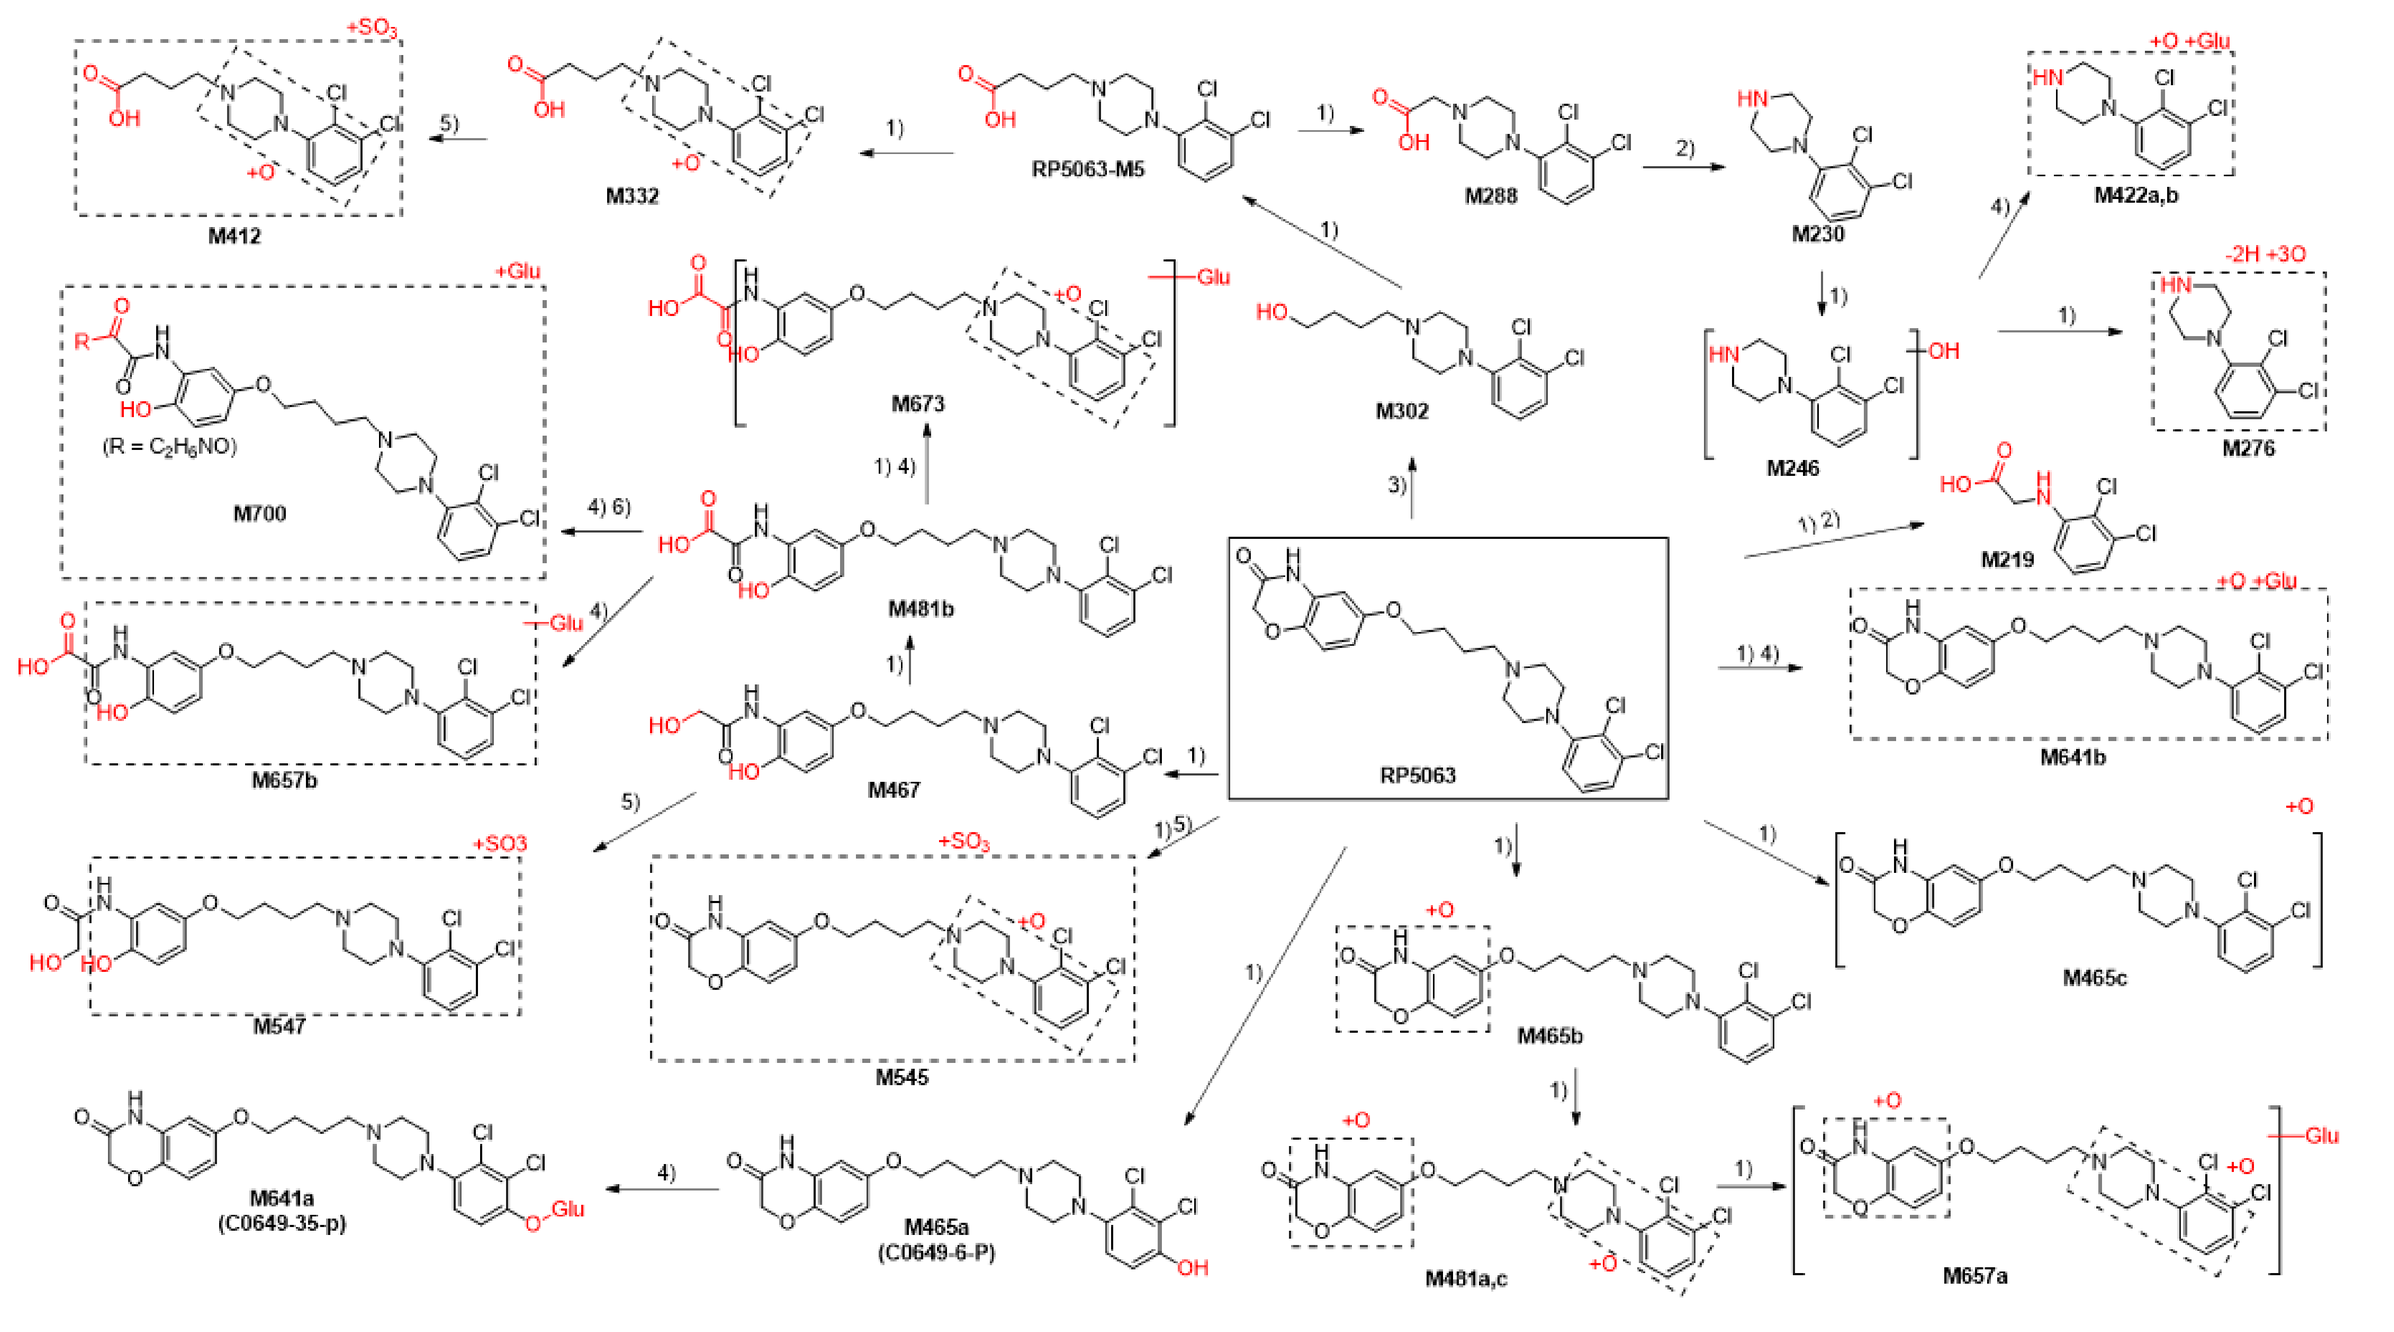
M412
M332
RP5063-M5
M288
M230
M422a,b
M700
M673
M302
M246
M276
M219
M481b
M657b
M467
RP5063
M641b
M547
M545
M465b
M465c
M641a (C0649-35-p)
M465a (C0649-6-P)
M481a,c
M657a

This study identified a total of 26 metabolites: M422a, M246, M332, M412, M230, M657a, M302, RP5081, M673, M481c, M641a, M641b, M481a, M288, M700, M465a, M467, M465b, M547, M545, M657b, M481b, M276, M422b, M465c, and M219. Oxidation, N- or O-dealkylation, reduction, sulfation, and glucuronidation were brilaroxazine's primary metabolic pathways. The formation of M641a represents the major metabolic pathway following a single oral dose at 20 mg/kg in male rats.

DISCUSSION

Oral brilaroxazine at 20 mg/kg in male SD rats underwent rapid and near complete absorption (at least 85%), extensive metabolism, and elimination as mostly metabolites via hepatobiliary excretion. Recovery of the dose was over 90% for both BDC and intact rats. The plasma PK profile involved a long elimination half-life (5.3 hr) for TRA and brilaroxazine, and a parent-to-total radioactivity based on a AUC_{last} of ~12%. Metabolites, M641a, RP5081, and M219, were the predominant entities circulating in rats.

A common metabolic pathway emerged from this work. Oxidation, N- or O-dealkylation, reduction, sulfation, and glucuronidation were the metabolic pathways observed, with the formation of M641a as the major metabolic pathway. The gut microflora likely appeared to convert the glucuronide metabolite, M641a, observed in the BDC rat bile to the mono-hydroxylated metabolite, M465a, which underwent fecal elimination in intact rats. Glucuronidase hydrolysis of M641a, producing M465a in a near-quantitative yield, confirmed this supposition.

CONCLUSION

This work defined the pharmacokinetics, metabolism, and excretion (PME) profile for a single oral dose of [^{14}C] brilaroxazine in SD rats. Mass balance was near quantitative with over 90% of the dose recovered. Bile was the predominant route of excretion in BDC rats and feces in intact rats. Also observed as major circulating metabolites in rats were circulating human metabolites, RP5081 and M219.[11] Furthermore, M465a was the major excreted metabolite in both humans[11] and rats. The emergent metabolic pathways for all species involved oxidation, N- or O-dealkylation with subsequent sulfation, and/or conjugation with glucuronic acid. These findings confirmed rats as a suitable rodent species for the brilaroxazine toxicological evaluation.

ACKNOWLEDGEMENTS & DISCLOSURES

Disclosures

Laxminarayan Bhat, PhD, and Seema R Bhat, MSc, are employees. Palaniappan Kulanthaivel, PhD, is a consultant for Reviva Pharmaceutical Holdings, Inc.

Acknowledgements :

John M. York, PharmD, MBA, of Akita Biomedical, Inc. provided editorial support, and Reviva Pharmaceuticals, Inc. funded this effort.

REFERENCES

1. Cantillon M, et al. *Clin Transl Sci*. 2018.
2. Cantillon M, et al. *Clin Transl Sci*. 2018
3. Cantillon M. et al. (2017). *Schizophr Res*. 189:126-133.
4. Bhat L, et al. *Society of Biologic Psychiatry*. 2023.
5. Bhat L, et al., *Medical Research Archives*. 2023.
6. Bhat L, et al. *Eur J Pharmacol*. 2017.
7. Bhat L, et al. *Eur J Pharmacol*. 2017.
8. Bhat L., et al., *Schizophrenia Int Res Soc*. 2024.
9. Bhat L, et al., *Am Soc for Clin Pharm and Therap*. 2024.
10. Cantillon M, et al. *Schizophrenia Research*. 2017.
11. Bhat L, et al., *J Pharmacol Exp Therap*. 2023.