Brilaroxazine Displays Limited Interaction with Clinically Relevant Drug Transporters

Laxminarayan Bhat*, Seema Bhat, and Palaniappan Kulanthaivel
Reviva Pharmaceuticals, Inc., 10080 N Wolfe Road, Suite SW3-200, Cupertino, California, USA

bit.ly/ASPETTransporters

ASPET Poster # 292 | May 17, 2024
ADDC Poster # 806 | May 19, 2024
Abstract ID # 129049

BACKGROUND

Brilaroxazine, a novel multimodal neuromodulator, belongs to a class of next-generation treatments for schizophrenia and comorbid conditions.[1-2] This novel multimodal dopamine (D) and serotonin (5-HT) receptor neuromodulator mitigates multiple inflammatory cytokines.[1-9] It specifically functions as a $D_{2/3/4}$ and 5-$HT_{1A/2A}$ partial agonist and a 5-$HT_{2B/7}$ antagonist with binding affinity for 5-HT_{2B} > D_2.[1-4]

This agent possesses an established efficacy, safety, and pharmacokinetic (PK) profile based on its Phase 1 and 2 clinical experience, including the Phase 2 REFRESH study.[1,2,10] It offers a differentiated pharmacological and safety profile over other antipsychotics.[1-10] Brilaroxazine's Phase 3 RECOVER study reflects a highly effective and well-tolerated treatment at the 50 mg daily dose and neuroinflammatory capabilities.[8,9]

Transporters influence drug absorption, distribution, and elimination, leading to PK and pharmacodynamic drug-drug interactions (DDIs) with co-administered inhibitors and/or inducers. Conversely, a drug can also modulate transporter functions, resulting in altered disposition of endogenous and exogenous substances. Regulatory agencies have recommended that sponsors should evaluate their investigational drugs both as substrates and inhibitors of clinically relevant transporters.

OBJECTIVE

This *in vitro* study's primary focus is to evaluate brilaroxazine's interaction potential with 11 clinically relevant drug transporters as a substrate and inhibitor. These included efflux transporters P-glycoprotein (P-gp), breast cancer resistance protein (BCRP), and bile salt export pump (BSEP); and uptake transporters multi-drug and toxin extrusion 1 (MATE1), MATE2-K, organic anion transporter 1 (OAT1), OAT3, organic cation transporter 1 (OCT1), OCT2, organic anion transporting polypeptide 1B1 (OATP1B1), and OATP1B3.

METHODS

Transporter Expressing Cell Lines

P-gp and BCRP substrate/inhibitor studies involved Madin-Darby canine kidney (MDCKII) epithelial cells (Sigma-Aldrich, St. Louis, MO). OATP1B1, OATP1B3, OAT1, OAT3, OCT1, OCT2, MATE1, and MATE2-K evaluation involved human embryonic kidney (HEK293) cells expressing these transporters, BSEP evaluation involved HEK293-derived human BSEP membrane vesicles, and mock HEK293 cells as controls (Corning Life Sciences, Bedford, MA).

Transporter Control Substrates and Inhibitors

The study employed standard inhibitors and substrates as controls (Table 1).

Table 1. Positive Substrates and Positive Control Inhibitors of Transporters

Transporter	Substrate	Inhibitor
P-gp	Digoxin (5 µM)	Ketoconazole (50 µM)
BCRP	Prazosin (5 µM)	Ko-143 (10 µM)
BSEP	TCA (10 µM)	Ketoconazole (100 µM)
OATP1B1	E17G (2 µM)	Rifampin (100 µM)
OATP1B3	E17G (5 µM)	Rifampin (100 µM)
OAT1	PAH (5 µM)	Diclofenac (200 µM)
OAT3	E3S (5 µM)	Probenecid (100 µM)
OCT1	Metformin (100 µM)	Verapamil (50 µM)
OCT2	Metformin (100 µM)	Amitriptyline (50 µM)
MATE1	Metformin (100 µM)	Cimetidine (100 µM)
MATE2-K	Metformin (100 µM)	Pyrimethamine (10 µM)

Brilaroxazine concentrations tested in the substrate assays were 3, 10, and 30 µM, and in the inhibition assays ranged from 0-100 µM.

Substrate and Inhibition Assays

The investigation performed all substrate and inhibition assays using established procedures. Concentration determination of brilaroxazine and control substrates involved using LC-MS/MS analysis (Sciex API 4000 and 4000 QTrap systems).

Data Analysis

The analysis utilized the following equations to calculate the Apparent Permeability (P_{app}) and Efflux Ratio (ER):

- $P_{app} = (dC_R/dt \times V_R)/(S \times C_{D,\,0hr})$
- $ER = P_{app}$ (B to A)/P_{app}(A to B)

Where dC_R/dt = rate of appearance of substrate or test article in the receiver chamber; S = surface area of the cell membrane; $C_{D,\,0hr}$ = initial drug concentration in the donor chamber; P_{app} (B to A) = apparent permeability from basal to apical membrane; P_{app} (A to B) = apparent permeability from apical to basal membrane.

Analysis calculated the IC_{50} using non-linear curve fittings with four parameters logistic equation:

$$y = d + \frac{a-d}{\left[(1+(\frac{x}{c})^b\right]}$$

RESULTS

Table 2

Table 2. Apparent Permeability (Papp) and Efflux Ratio (ER) of Brilaroxazine in P-gp and BCRP Expressed MDCKII Cell Monolayers

Transporter	Substrate	Concentration (mM)	(-) Inhibitor[1] P_{app} (10^{-6} cm/sec) A to B	(-) Inhibitor[1] P_{app} (10^{-6} cm/sec) B to A	(-) Inhibitor[1] ER	(+) Inhibitor[1] P_{app} (10^{-6} cm/sec) A to B	(+) Inhibitor[1] P_{app} (10^{-6} cm/sec) B to A	(+) Inhibitor[1] ER
P-gp	Brilaroxazine	3	0.24	2.16	9.18	2.71	5.12	1.89
		10	0.99	3.51	3.54	3.18	5.37	1.69
		30	2.76	4.72	1.71	4.55	6.39	1.40
	Digoxin	5	0.65	6.37	9.73	1.83	2.18	1.20
BCRP	Brilaroxazine	3	0.54	2.34	4.33	1.03	2.57	2.50
		10	1.46	2.22	1.52	2.20	4.10	1.86
		30	3.03	4.30	1.42	3.51	5.91	1.68
	Prazosin	5	0.82	15.89	19.40	9.09	10.35	1.14

[1] The investigation utilized ketoconazole as the P-gp inhibitor and Ko 143 as the BCRP inhibitor.

In P-gp-expressed MDCKII cells, the ERs of brilaroxazine were 9.18, 3.54 and 1.71 at 3, 10 and 30 µM, respectively. The ER decreased by >50% in the presence of a P-gp inhibitor, ketoconazole, indicating that brilaroxazine is a substrate of P-gp. Similarly, in BCRP-expressed MDCKII cells, the ERs of brilaroxazine were 4.33, 1.52 and 1.42 at 3, 10 and 30 µM, respectively, suggesting that brilaroxazine is a substrate of BCRP.

Evaluations with all other transporters, OATP1B1, OATP1B3, OAT1, OAT3, OCT1, OCT2, MATE1, MATE2-K and BSEP, failed to demonstrate any significant evidence of substrate interaction with brilaroxazine.

Table 3

Table 3. Inhibition of Transporters by Brilaroxazine

Transporter	IC50 (µM)	Substrate
P-gp	5.7	Digoxin (5 µM)
BCRP	12.3	Prazosin (5 µM)
BSEP	46.9	TCA (10 µM*)
OATP1B1	9.1	E17G (2 µM)
OATP1B3	23.6	E17G (5 µM)
OAT1	>100	PAH (5 µM)
OAT3	58.4	E3S (5 µM)
OCT1	16.3	Metformin (100 µM)
OCT2	56.4	Metformin (100 µM)
MATE1	0.5	Metformin (100 µM)
MATE2-K	7.5	Metformin (100 µM)

*Concentration of cholic acid, converted to TCA *in situ*

Brilaroxazine inhibited OATP1B1, OATP1B3, OAT1, OAT3, OCT1, OCT2, MATE2-K, BSEP, P-gp, and BCRP with IC_{50} values ranging from 5.7 µM to >100 µM. MATE1 was the only transporter with an IC_{50} of <1 µM.

Figure 1

Figure 1. Brilaroxazine as Inhibitor of Transporters

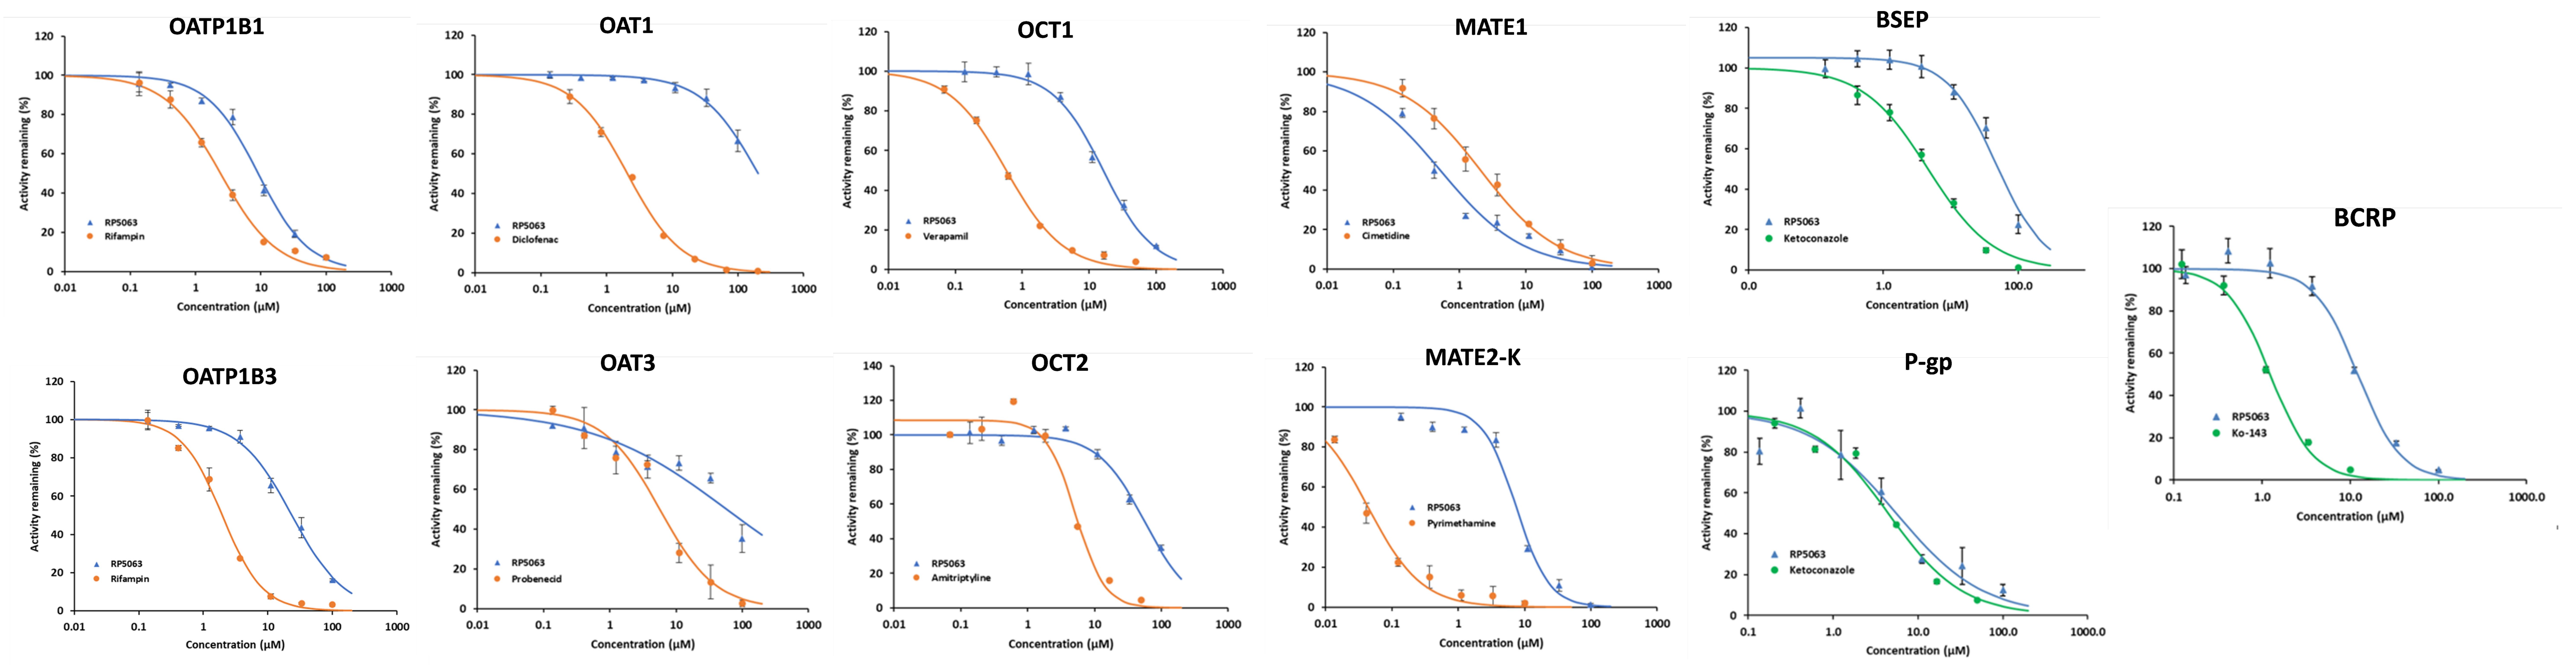
OATP1B1
OAT1
OCT1
MATE1
BSEP
BCRP
OATP1B3
OAT3
OCT2
MATE2-K
P-gp
Activity remaining (%)
Concentration (µM)
RP5063
Rifampin
Diclofenac
Verapamil
Cimetidine
Ketoconazole
Ko-143
Probenecid
Amitriptyline
Pyrimethamine

Inhibition curves for OAT1B1, OAT1B3, OAT1, OAT3, OCT1, OCT2, MATE1, MATE2-K, BSEP, P-gp and BCRP by brilaroxazine and prototypical inhibitors. All transporter inhibition studies utilized bilaroxazine concentrations of 0, 0.1, 0.4, 1.2, 3.7, 11.1, 33.3 and 100 µM. For control inhibitors, a concentration range of 0-10 µM was used for pyrimethamine (MATE2-K) and Ko-143 (BCRP), 0-50 µM for verapamil (OCT1), amitriptyline (OCT2), and ketoconazole (P-gp), 0-100 µM for rifampin (OATP1B1 and OATP1B3), probenecid (OAT3), cimetidine (MATE1), and ketoconazole (BSEP) and 0-200 µM for diclofenac (OAT1).

DISCUSSION

Significance of Transporter Inhibition by Brilaroxazine

The scientific and regulatory community recognizes that drug transporters—solute carriers and ATP-binding cassette types—can affect drug PK, safety, and efficacy.[12-14] While drug-metabolizing enzyme activity occurs primarily in the liver and small intestines, transporter expression occurs in tissues throughout the whole human body.[13,14] Because they can govern the access of endogenous and exogenous substances to various sites in the body, it is essential to evaluate transporter-mediated interactions with new pharmaceutical compounds.[14]

Regulatory agencies have recommended that sponsors evaluate their investigational drugs as substrates and/or inhibitors of clinically relevant transporters. Accordingly, this work sought to address this requirement[12-14] and evaluated 11 clinically relevant transporters.

In vitro, brilaroxazine is a substrate of efflux transporters P-gp and BCRP. Co-administration with strong inhibitors/inducers of these transporters may increase/decrease its systemic exposure. In a clinical drug interaction study with itraconazole, a potent inhibitor of P-gp and BCRP, the study observed no altered PKs of brilaroxazine, indicating that the *in vitro* data did not translate *in vivo* in humans.[11]

Brilaroxazine is not a substrate of OATP1B1, OATP1B3, OAT1, OAT3, OCT1, OCT2, MATE1. One should not expect substrate interactions with MATE2-K and BSEP and drug interactions when brilaroxazine is co-administered with inhibitors or inducers of the above transporters.

Brilaroxazine inhibited P-gp and BCRP with IC_{50} values of 5.7 and 12.3 µM. Based on FDA[15] and EMA[16] guidance documents, a DDI is possible due to inhibition of P-gp and BCRP at the gastrointestinal (GI) tract when I_{gut}/IC_{50} is ≥10, where I_{gut} is the luminal concentration of the interacting drug calculated as the dose/250 mL or, if low solubility, the maximum possible concentration at the pH range of the GI tract (EMA only) and IC_{50} is the half-maximal inhibitory concentration. For P-gp and BCRP inhibition, the I_{gut}/IC_{50} values were >10 based on the highest brilaroxazine therapeutic dose of 50 mg, suggesting that there may be a potential for DDI in the GI tract. However, based on the I_1/IC_{50}, where I_1 is the $C_{max,\,ss}$ at the highest therapeutic dose of 50 mg, and IC_{50} is the half-maximal inhibitory concentration, systemic DDI is unlikely to occur.

Brilaroxazine inhibited the hepatic uptake transporters OATP1B1, OATPIB3 and OCT1 with IC_{50} values of 9.1, 23.6 and 16.3 µM, respectively. The R values estimated based on the steady-state exposure at the highest therapeutic dose of 50 mg suggest that DDI is unlikely when brilaroxazine is co-administered with substrate drugs of OATP1B1, OATP1B3 and OCT1.[15] Brilaroxazine inhibited the hepatic efflux transporter BSEP with an IC_{50} of 46.9 µM. Since the 50-fold unbound $C_{max,\,ss}$ (estimated liver concentration) is well below the IC_{50} value, it is unlikely that brilaroxazine will inhibit BSEP clinically.[16]

Brilaroxazine inhibited the renal transporters OAT1, OAT3, OCT2, MATE1 and MATE2-K with IC_{50} values of >100, 58.4, 56.4, 0.5 and 7.5 µM. Based on the $I_{max,u}/IC_{50}$ (<0.1) at the highest therapeutic dose of 50 mg, DDI is unlikely when brilaroxazine is co-administered with substrate drugs of the above transporters.[16]

CONCLUSION

This study evaluated *in vitro* the interaction between brilaroxazine and 11 clinically relevant transporters. The results found that brilaroxazine displays limited interaction with clinically relevant drug transporters. Such findings indicate that in most cases, the potential for transporter-mediated DDI appears to be unlikely at the highest therapeutic dose. Further work in the clinic will help to confirm these *in vitro* findings.

ACKNOWLEDGEMENTS & DISCLOSURES

Disclosures:

Laxminarayan Bhat, PhD and Seema R Bhat, MSc are employees. Palaniappan Kulanthaivel, PhD is a consultant for Reviva Pharmaceutical Holdings, Inc.

Acknowledgements:

John M. York, PharmD, MBA, of Akita Biomedical, Inc. provided editorial support, and Reviva Pharmaceutical Holdings, Inc. funded this effort.

REFERENCES

1. Cantillon M, et al. *Clin Transl Sci*. 2018.
2. Cantillon M, et al. *Clin Transl Sci*. 2018.
3. Cantillon M. et al. (2017). *Schizophr Res*. 189:126-133.
4. Bhat L, et al. *Society of Biologic Psychiatry*. 2023.
5. Bhat L, et al., *Medical Research Archives*. 2023.
6. Bhat L, et al. *Eur J Pharmacol.* 2017.
7. Bhat L, et al. *Eur J Pharmacol.* 2017.
8. Bhat L., et al., *Schizophrenia Int Res Soc.* 2024.
9. Bhat L, et al., *Am Soc for Clin Pharm and Therap.* 2024.
10. Cantillon M, et al. *Schizophrenia Research.* 2017.
11. Bhat L, et al., *J Pharmacol Exp Ther.* 2023.
12. Yu J and Ragueneau-Majlessi, *Clin Transl Sci.* 2020.
13. Elsby R, et al. *Exp Opin on Drug Metab & Tox.* 2022.
14. Fu S, et al. *Medicine in Drug Discovery*. 2021.
15. FDA Guidance for Industry. *In vitro* drug interaction studies – cytochrome P450 enzyme- and transporter-mediated drug interactions, January 2020.
16. EMA Guideline on the investigation of drug interactions, 2012.